Population PK (PopPK) and Pharmacokinetic/Pharmacodynamic (PK/PD) Analysis of Avexitide in Individuals with Post-Bariatric Hypoglycemia

Colleen Craig, MD[1*]; Kelly Fox, MD[2*]; Machelle Manuel, PhD[2*]

[1]Stanford University School of Medicine, Department of Medicine, Stanford, California, USA; [1]Amylyx Pharmaceuticals, Inc., Cambridge, Massachusetts, USA. *Potential conflict of interest may exist. Refer to the Disclosures section.

ENDO TOPICAL CATEGORY
DIABETES AND VASCULAR DISEASE

SAT-559

AMYLYX®

BACKGROUND

- Post-bariatric hypoglycemia (PBH) is a chronic condition characterized by hyperinsulinemic hypoglycemia that develops in some individuals who have undergone bariatric surgery[1-6]
- PBH is believed to be caused by changes in hormonal and glycemic patterns, including an excessive glucagon-like peptide-1 (GLP-1) response, as a result of altered nutrient transit post-surgery[1-4]
- Endogenous GLP-1 levels fluctuate throughout the day,[5,7-9]
 - In healthy, bariatric surgery-naive individuals, fasting GLP-1 concentrations are generally low, typically <10 pM, with levels rising 2- to 3-fold after eating, peaking 20 to 30 minutes after the meal
 - GLP-1 levels exhibit a diurnal rhythm with highest levels typically observed during the daytime and levels returning to baseline between meals and overnight
 - In PBH, an exaggerated form of these GLP-1 responses is believed to be a central pathway causing inappropriately elevated insulin levels, leading to persistent, recurrent, and debilitating hypoglycemia **(Figure 1)**

FIGURE 1. Illustrative Example of Daily GLP-1 Levels

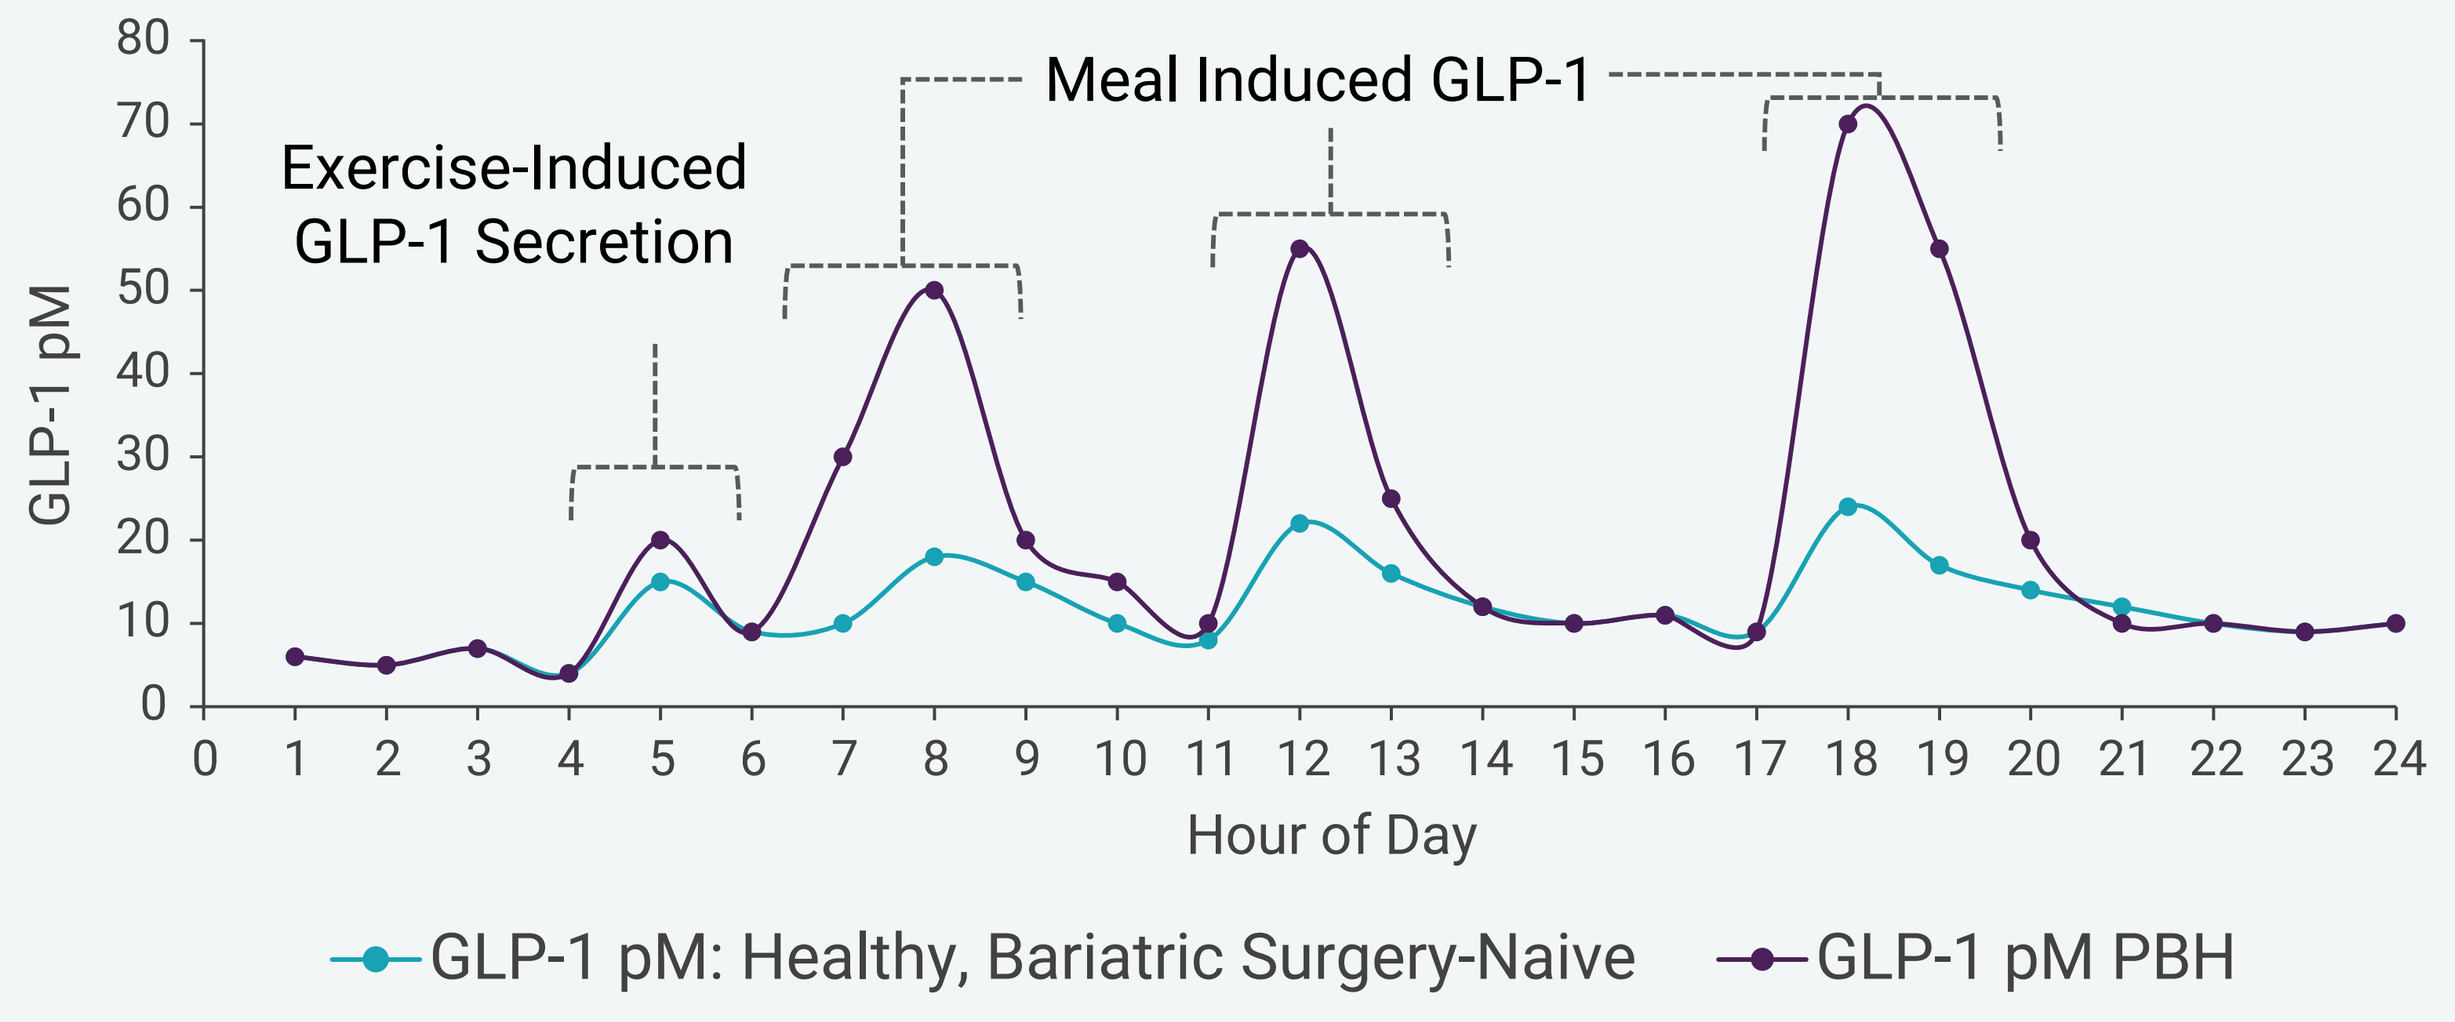
Exercise-Induced GLP-1 Secretion
Meal Induced GLP-1
GLP-1 pM
Hour of Day
GLP-1 pM: Healthy, Bariatric Surgery-Naive
GLP-1 pM PBH

- Avexitide is an investigational, first-in-class glucagon-like peptide-1 (GLP-1) receptor antagonist designed to block the effect of excessive GLP-1, targeting a central pathway of PBH pathophysiology to mitigate hypoglycemia by decreasing insulin secretion and normalizing glucose levels[1-4]

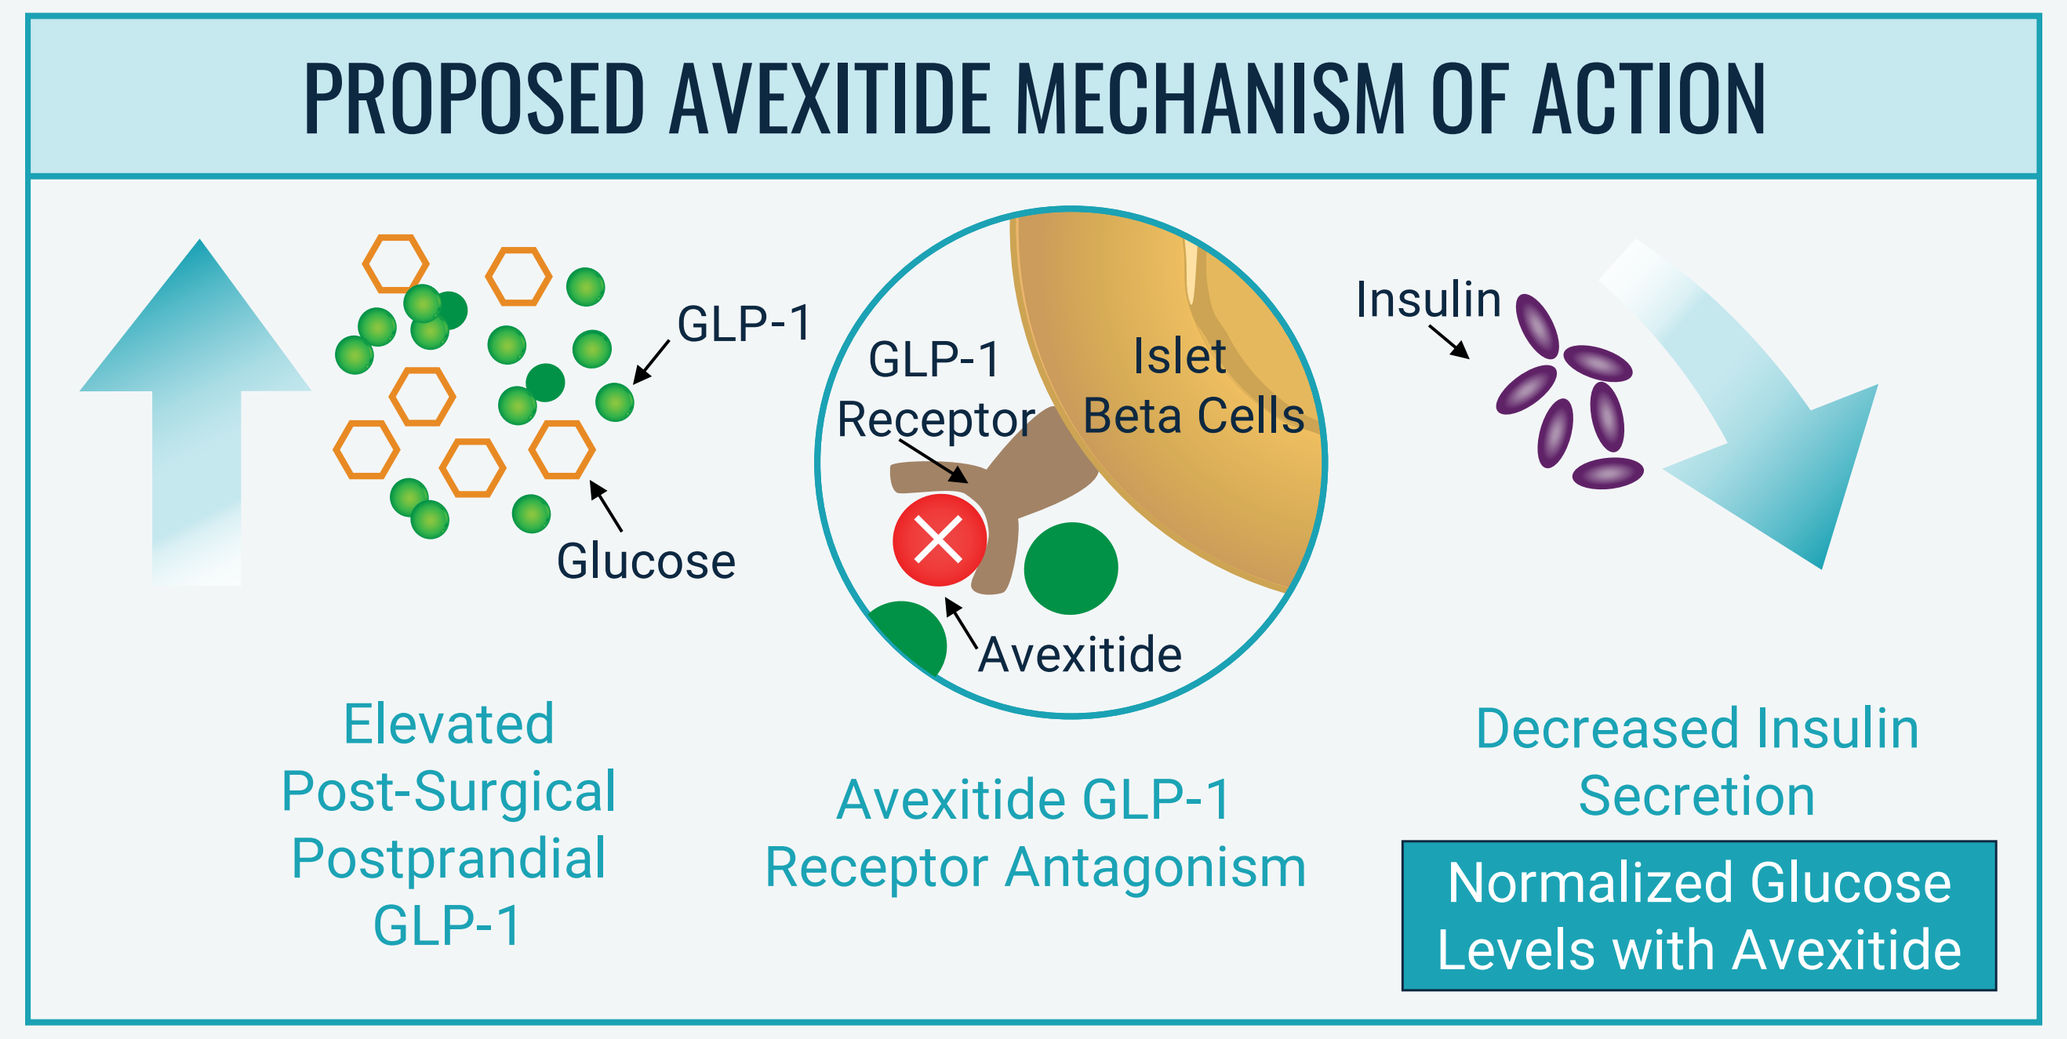
PROPOSED AVEXITIDE MECHANISM OF ACTION
GLP-1
Glucose
GLP-1 Receptor
Islet Beta Cells
Avexitide
Insulin
Elevated Post-Surgical Postprandial GLP-1
Avexitide GLP-1 Receptor Antagonism
Decreased Insulin Secretion
Normalized Glucose Levels with Avexitide

OBJECTIVE

Develop a population pharmacokinetic (PopPK) model for avexitide in people with PBH and evaluate the pharmacokinetic profile and pharmacodynamic response of avexitide to help establish the relationship between plasma drug concentrations and therapeutic effects in PBH

Study 1: *In Vitro* Potency Studies
Avexitide Inhibited GLP-1 Receptor Activity

METHODS

- **Assay Technology:** Homogenous Time Resolve Fluoresence (HTRF) cAMP kit (Revvity)
 - Competitive immunoassay between cAMP produced by cells and cAMP labelled with a fluorophore (d2)
- **Cell System:** hGLP-1R CHO-K1 cells (2,000 cells/well) in suspension, directly thawed from frozen assay-ready stocks from Euroscreen
- **Assay Approach:** 30-minute incubation of 5 µL of cells with 2.5 µL of avexitide at increasing concentrations; 2.5 µL of GLP-1 at three different concentrations, including at its EC90 of 0.02 nM; and 5 µL of each fluorophore

RESULTS

- Avexitide had an IC50 of approximately 20-30 nM (70-100 ng/mL) in the presence of significant levels of GLP-1 **(Figure 2)**
- Avexitide had an IC90 of approximately 100 nM (350 ng/mL) in the presence of significant levels of GLP-1
- Limited difference was found between IC50 and IC90 with increasing GLP-1 concentrations

FIGURE 2. Avexitide *In Vitro* Potency Screen

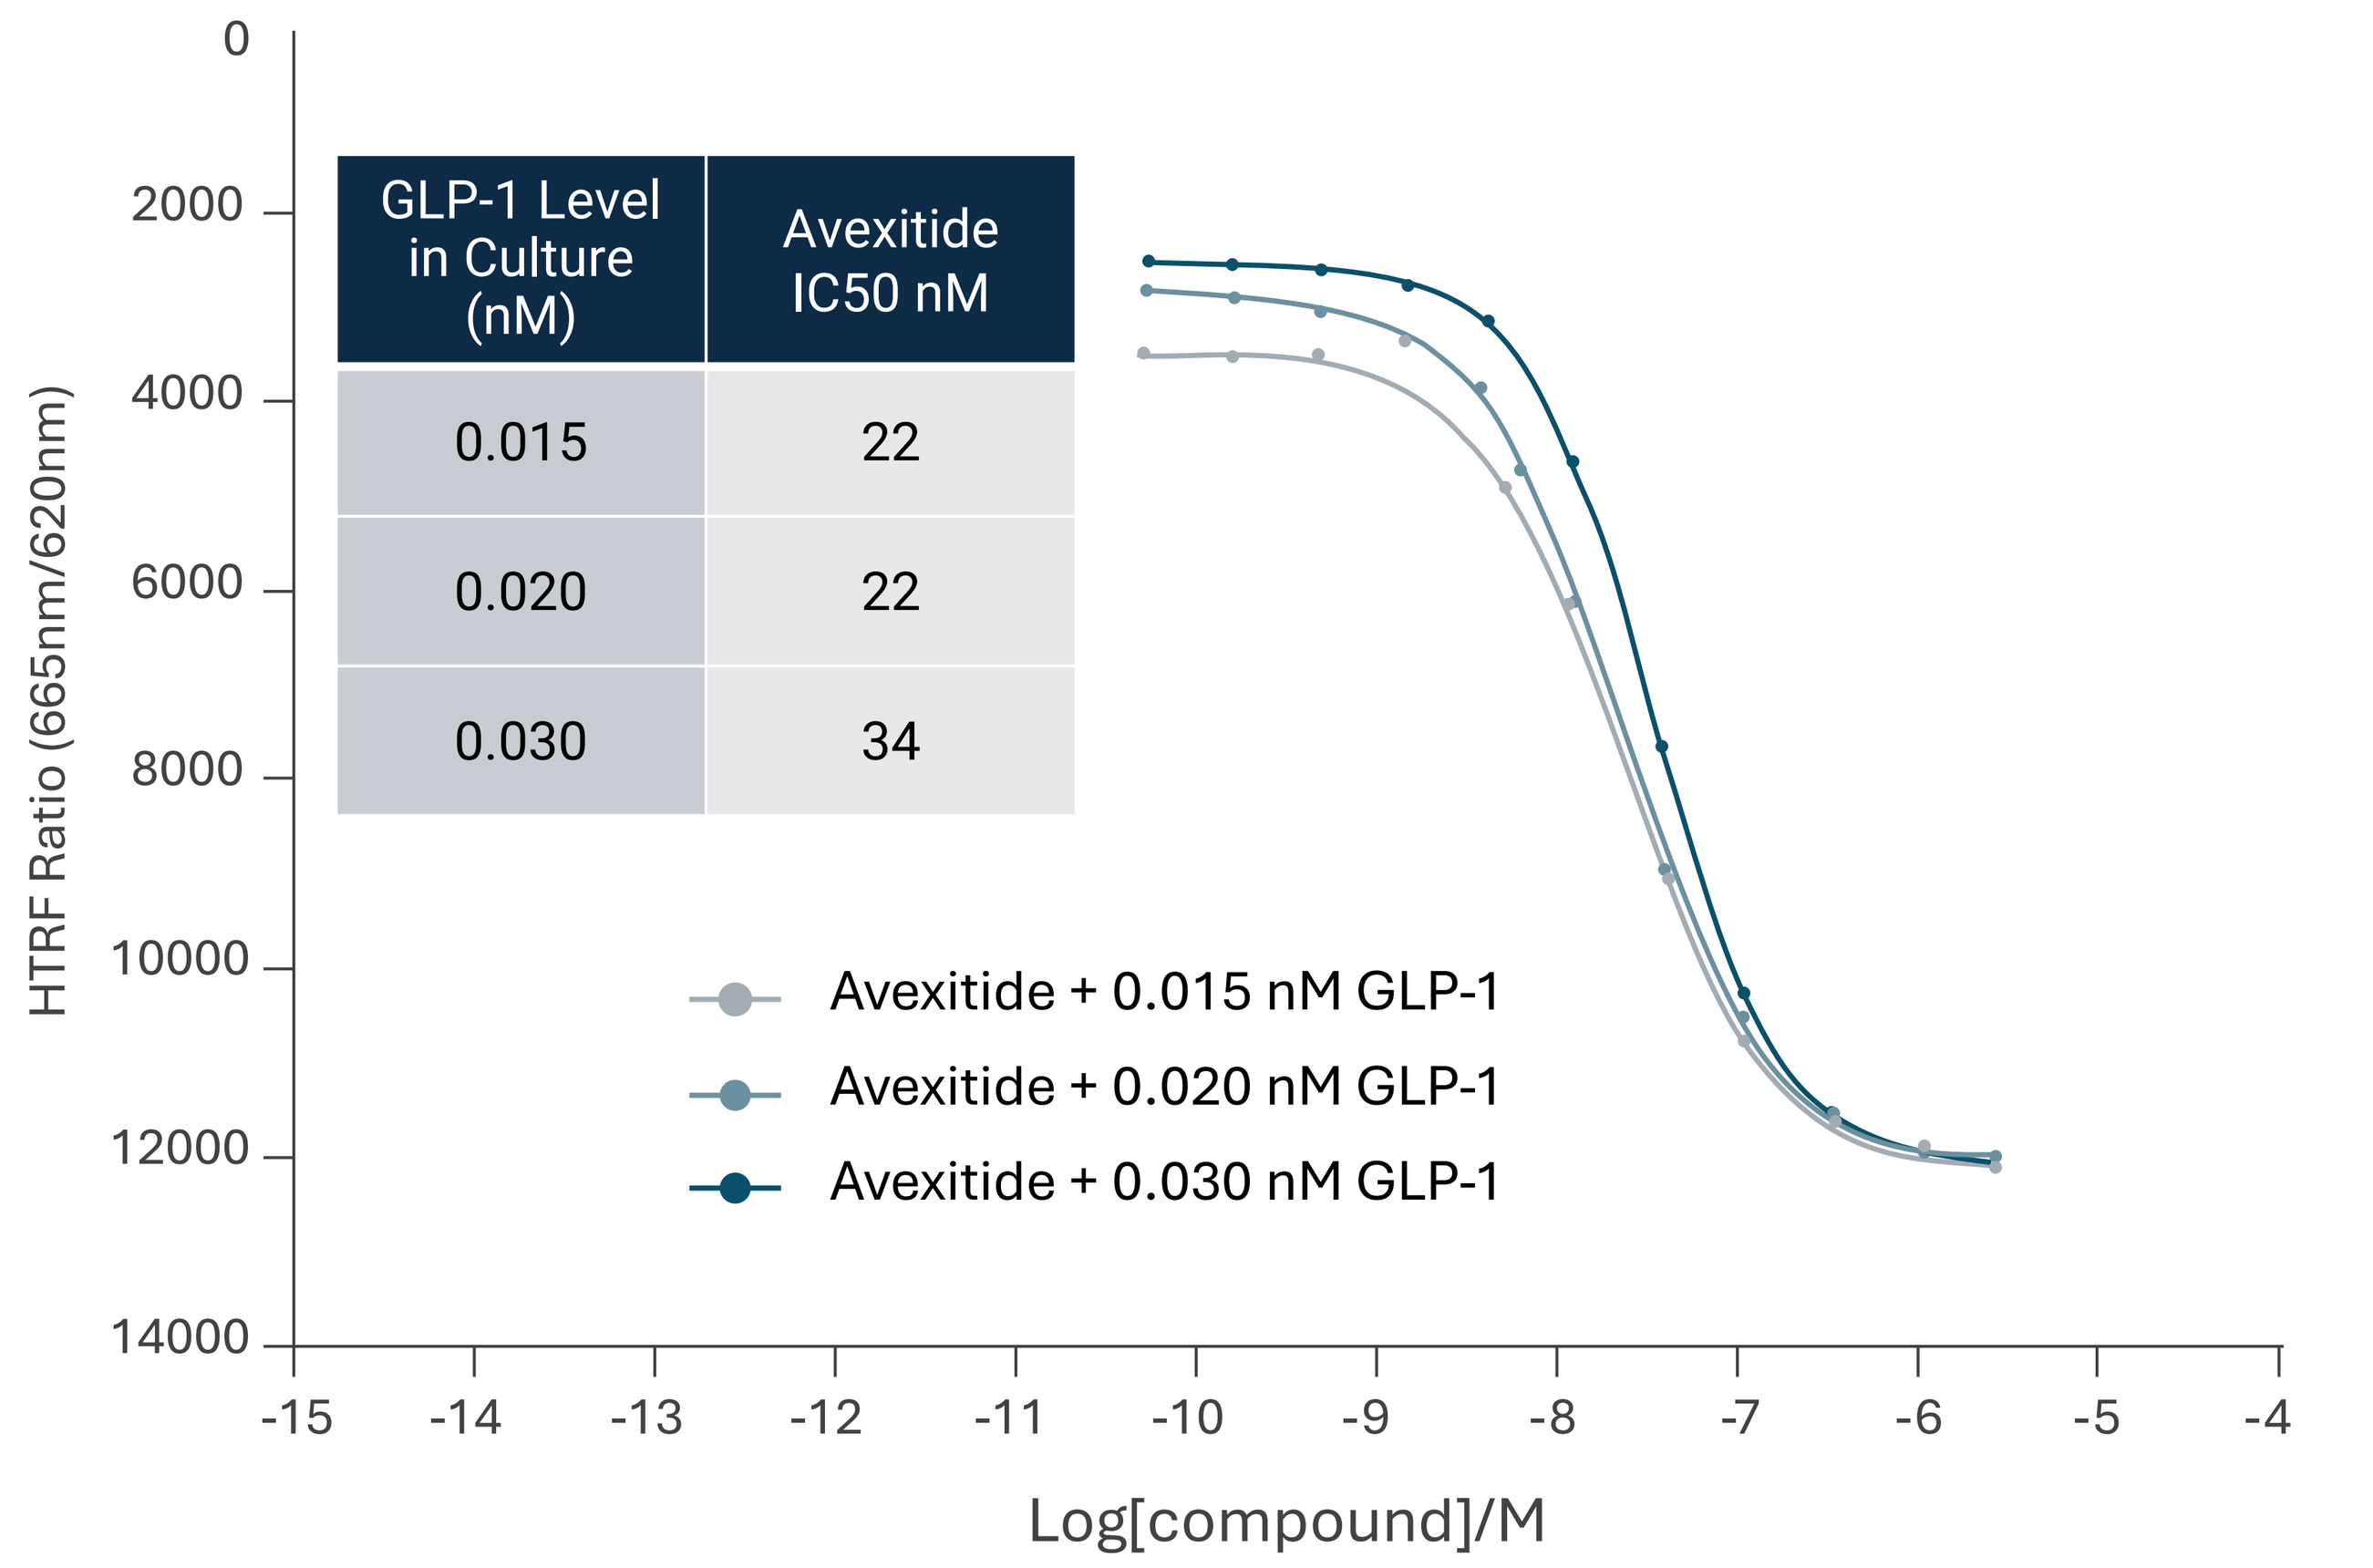

GLP-1 Level in Culture (nM)	Avexitide IC50 nM
0.015	22
0.020	22
0.030	34

HTRF Ratio (665nm/620nm)
Log[compound]/M
Avexitide + 0.015 nM GLP-1
Avexitide + 0.020 nM GLP-1
Avexitide + 0.030 nM GLP-1

Study 2: PopPK Analysis
90 mg Avexitide Exceeded IC50 for 24H and IC90 during Waking Hours

METHODS

- The PK of avexitide in plasma was evaluated in 58 individuals with 1,473 samples from two studies *(EIG-EXD-001/PREVENT and EIG-EXD-002)*
- A nonlinear mixed effects modeling approach with the first-order conditional estimation (FOCE) method in NONMEM 7, version 7.3 (ICON, Maryland) was used for the PopPK analysis
- The impact of baseline covariates, including age, weight, BMI, gender, race, ALT, AST, bilirubin, and EGFR on the PK of avexitide were investigated
 - Covariates were selected using a forward addition and backward elimination method (based on the significance levels of $p<0.01$ and $p<0.001$, respectively)

RESULTS

- The structural PopPK model that best described the avexitide data was a one-compartment model with sequential zero- then first-order absorption, first-order elimination from the central compartment
 - Body weight effect on clearance and central volume of distribution were identified as two significant covariates for avexitide population PK
 - PopPK model successfully validated based on goodness-of-fit plots, visual predictive check (VPC), and other diagnostics
- Typical avexitide PK profiles at steady-state following 60 and 90 mg daily dosing simulated based on PK parameters estimated from the final avexitide PopPK model are shown in **Figure 3** and concentration vs. time curves for avexitide in healthy individuals in EIG-EXD-002 are shown in **Figure 4**
- 90 mg avexitide exceeded IC50 for 24 hours and maintained IC90 from morning to midnight

FIGURE 3. Simulated Avexitide (AVX) Plasma Concentration Following Daily Treatment in PBH

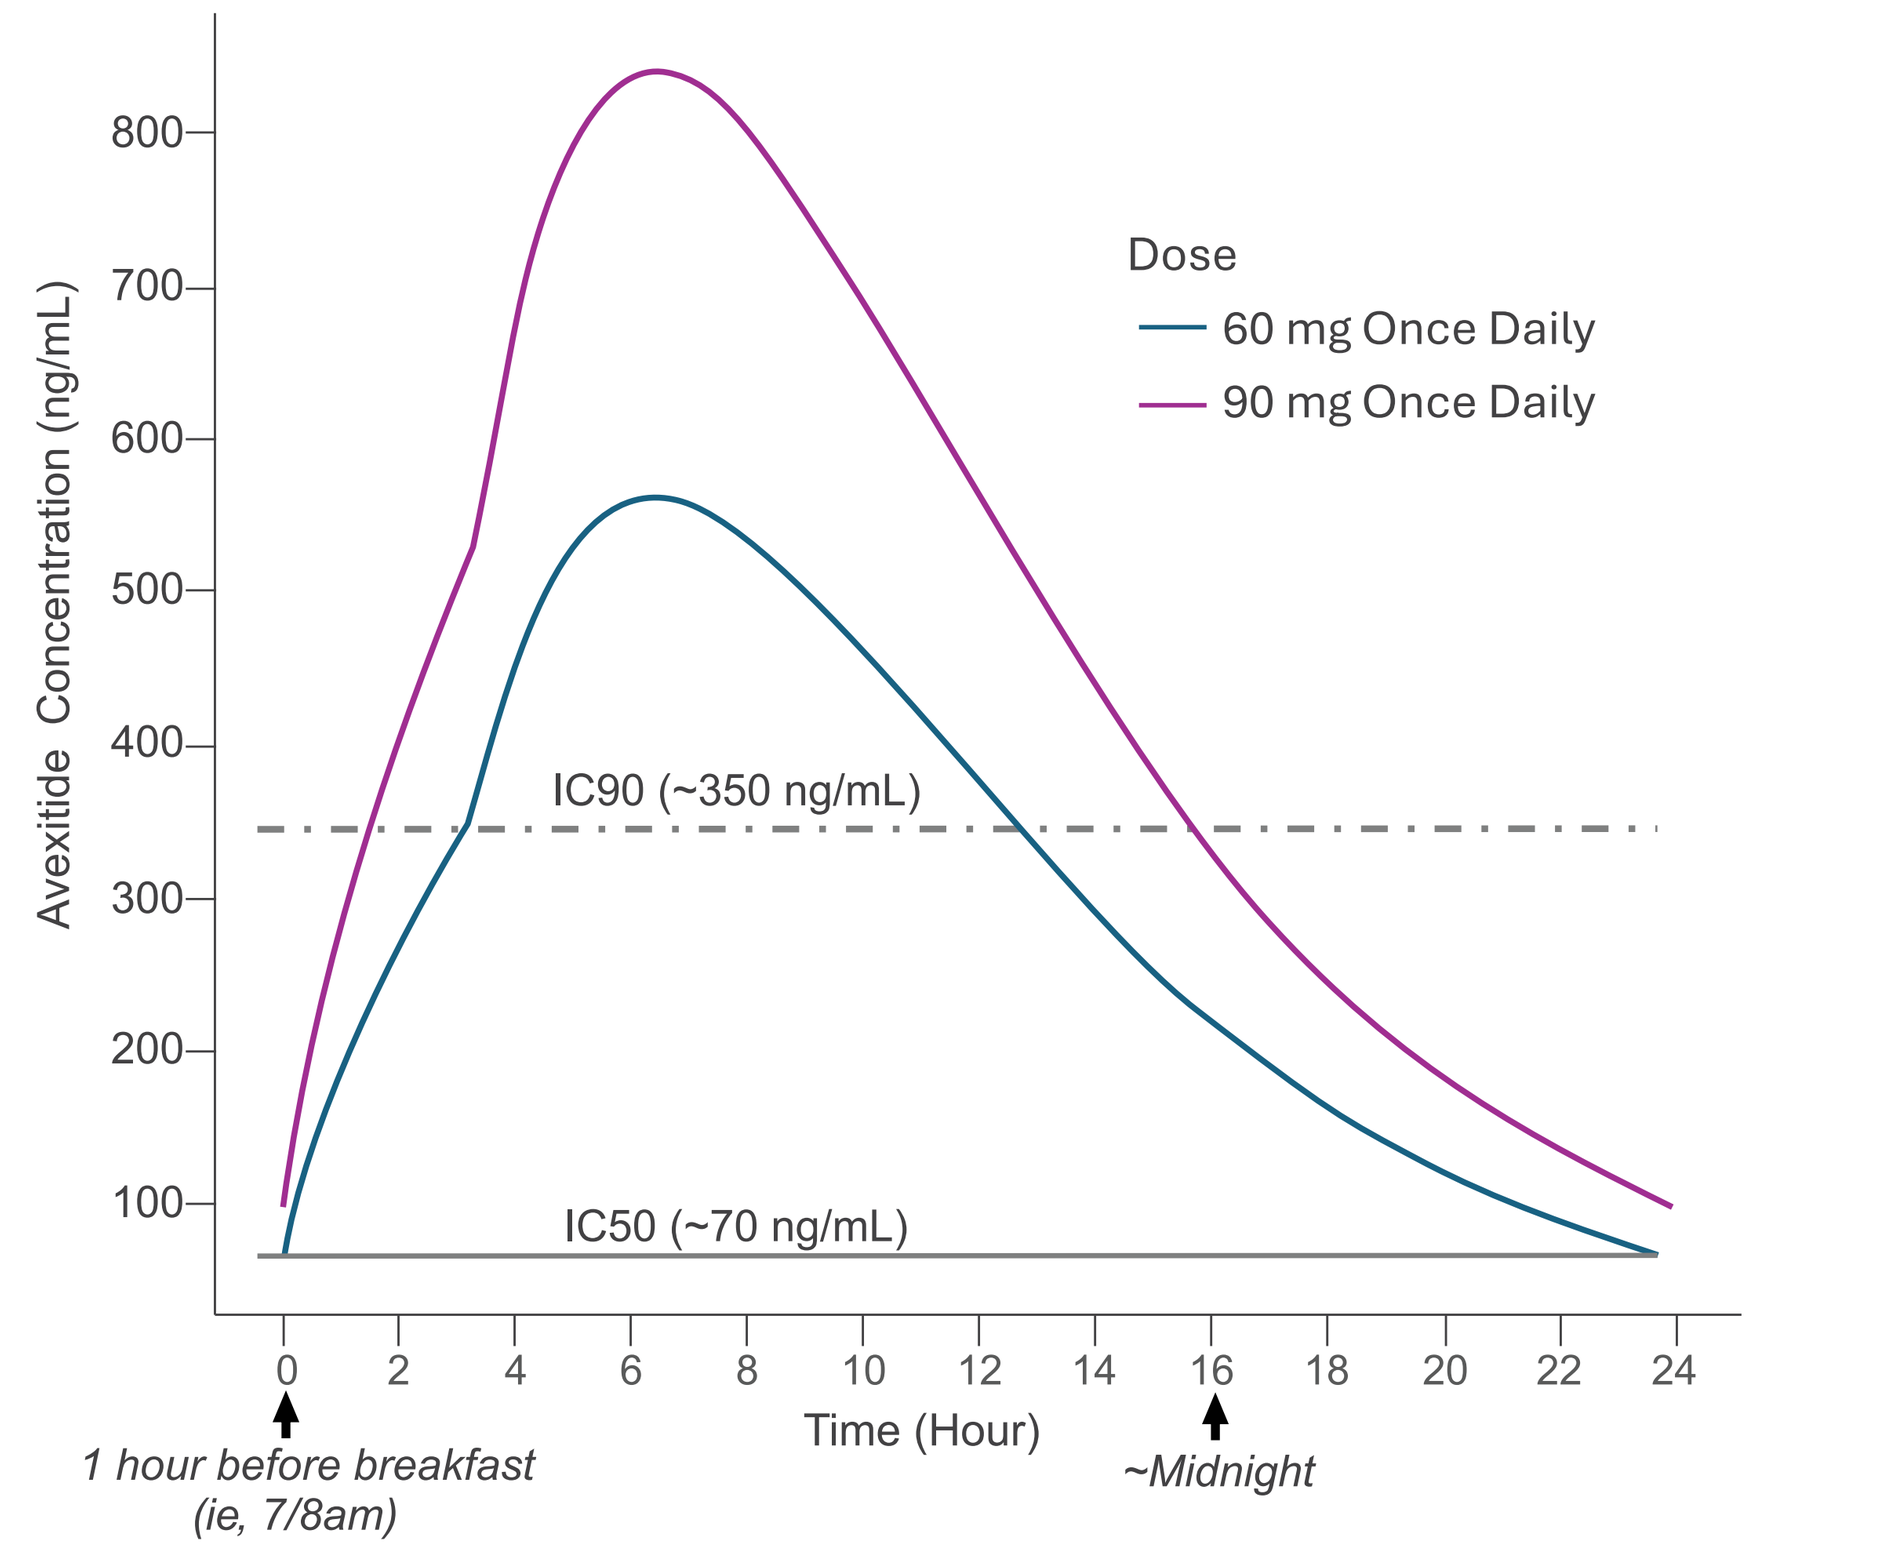
Avexitide Concentration (ng/mL)
Dose
60 mg Once Daily
90 mg Once Daily
IC90 (~350 ng/mL)
IC50 (~70 ng/mL)
Time (Hour)
1 hour before breakfast (ie, 7/8am)
~Midnight

FIGURE 4. Mean ± SD AVX Plasma Concentration in Healthy Individuals in EIG-EXD-002

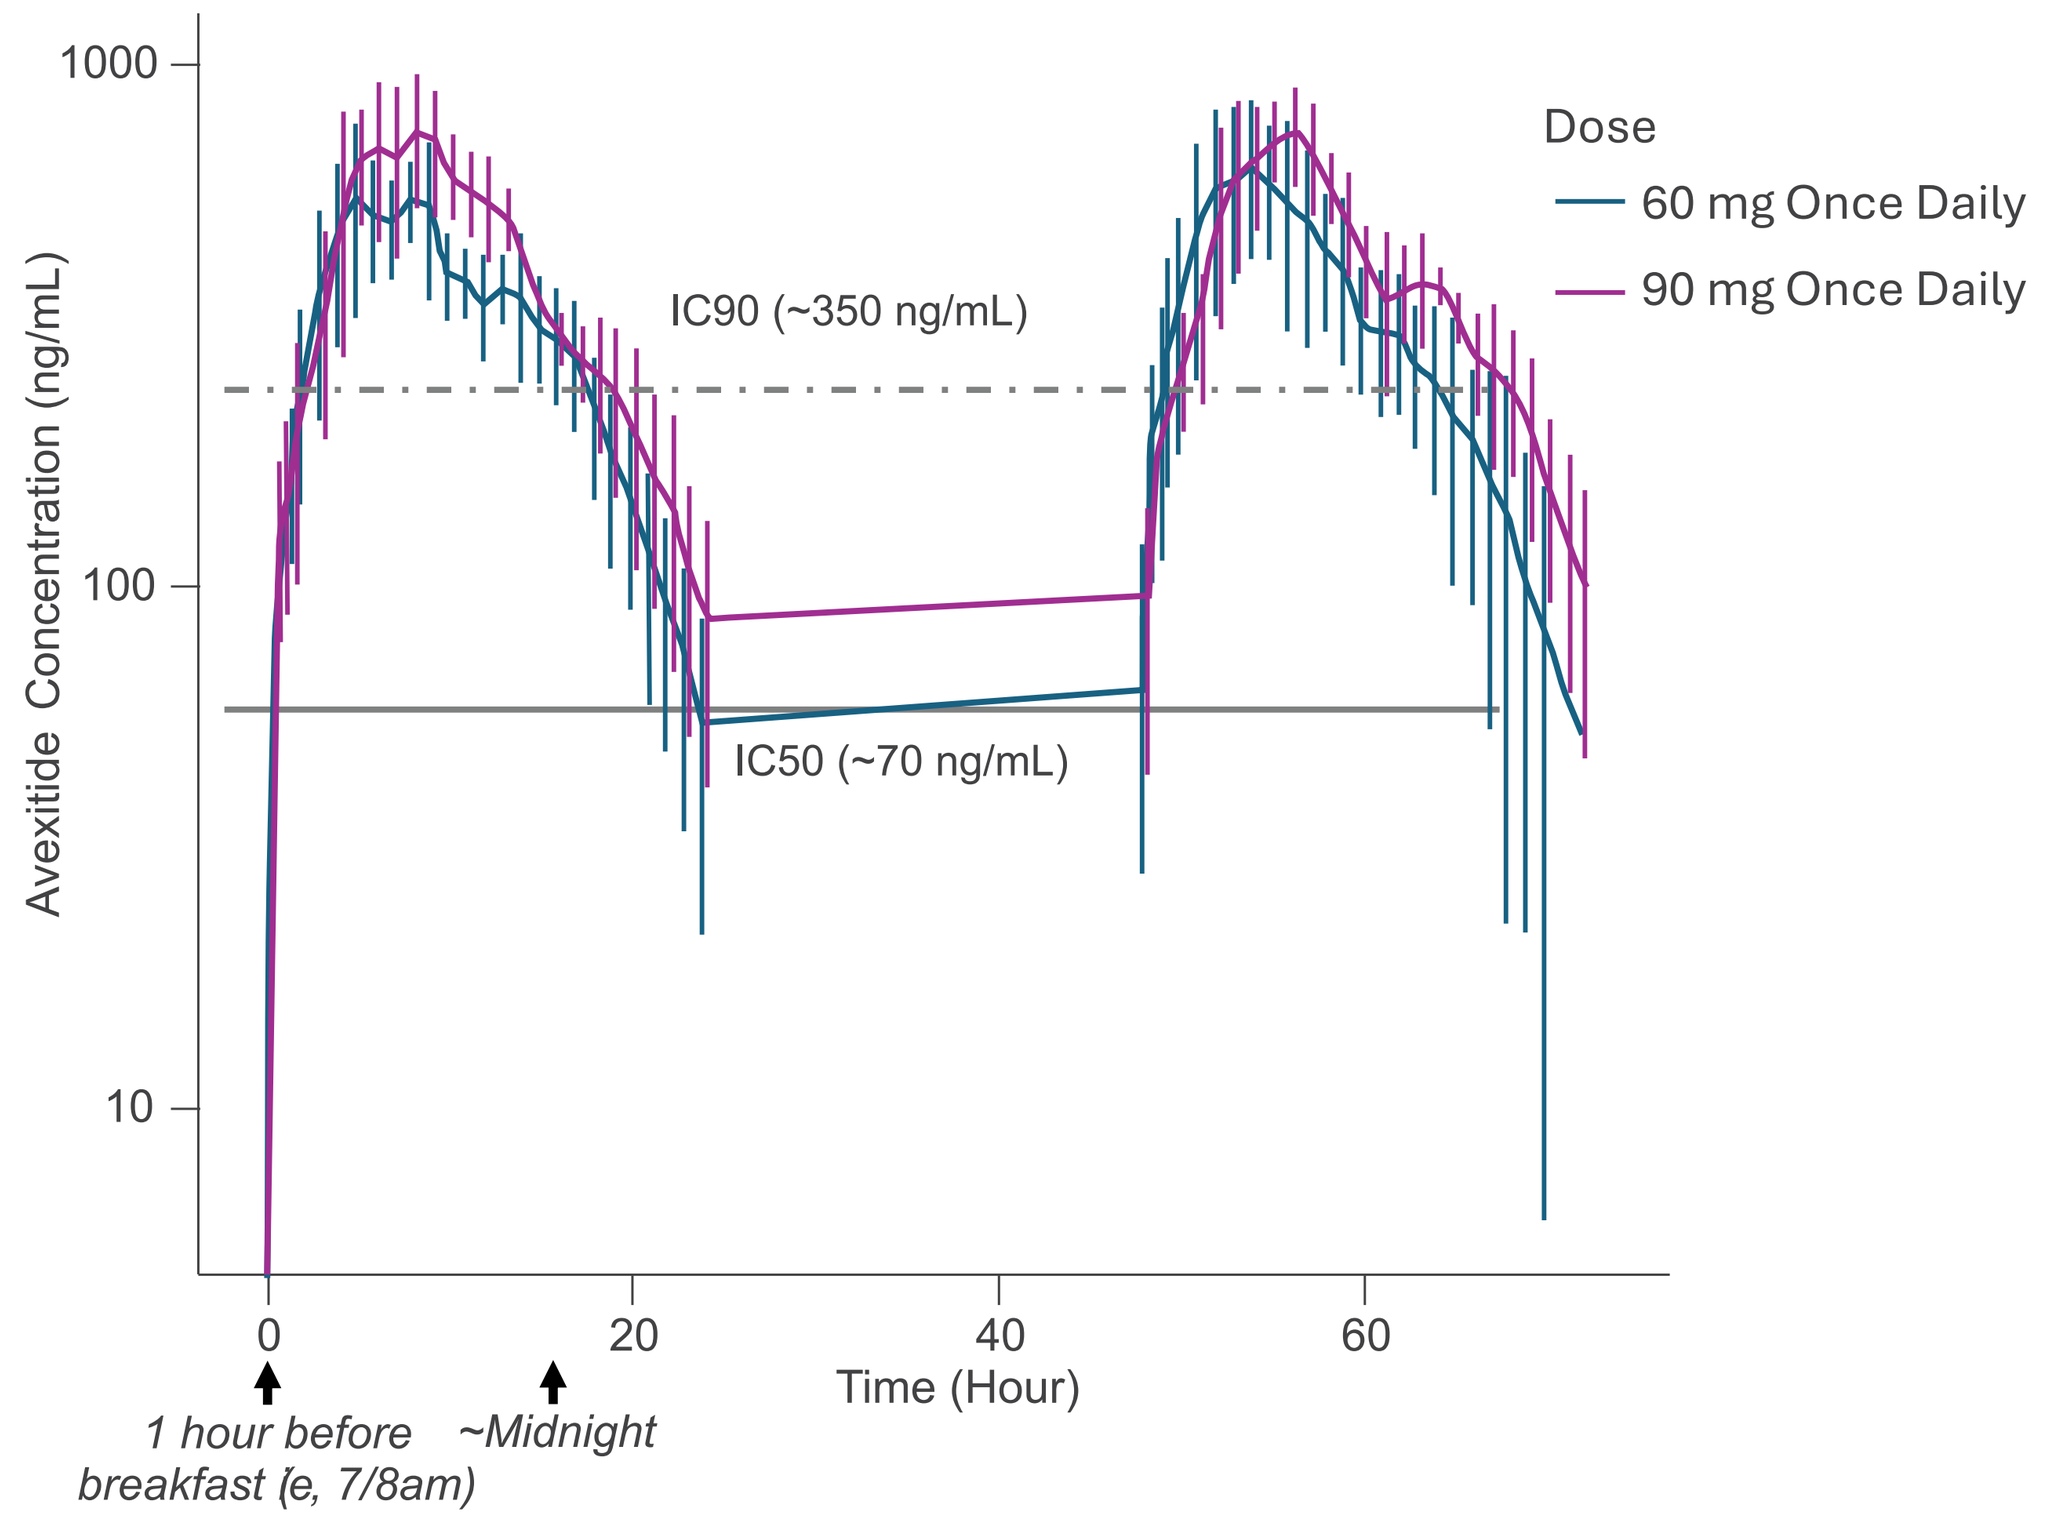
Avexitide Concentration (ng/mL)
Dose
60 mg Once Daily
90 mg Once Daily
IC90 (~350 ng/mL)
IC50 (~70 ng/mL)
Time (Hour)
1 hour before breakfast (e, 7/8am)
~Midnight

Note: IC_{50} and IC_{90} lines are estimated values based on Study 1 and are overlaid on data from Study 2. These estimates should be interpreted with appropriate caution.

CONCLUSIONS

- Avexitide is an inhibitor of GLP-1 receptor activity
- Avexitide 90 mg once daily maintains concentration above IC50 for 24 hours
- Avexitide 90 mg once daily maintained IC90 from morning to midnight, the most relevant hours for hypoglycemic events given GLP-1 diurnal patterns
- Findings are consistent with Phase 2b CGM data, which demonstrated similar hypoglycemic event rate reduction during day and night-time hours at the 90 mg once daily dose
- In addition to the Phase 2b results, PK data further support the rationale for and appropriateness of the 90 mg once daily dosing regimen in the Phase 3 LUCIDITY trial

lucidity

SCAN TO LEARN MORE ABOUT THE PHASE 3 LUCIDITY TRIAL

CONTACT INFO: KELLY FOX, MD
Kelly_Fox@amylyx.com

Avexitide is an investigational drug and has not been approved for use by any health authority (e.g., the FDA and EMA).

Acknowledgements

We gratefully acknowledge Gubra for their valuable data contributions and scientific collaboration that supported the *in vitro* potency analysis. The authors would also like to thank the individuals who provided data supporting this analysis.

Disclosures

CC reports consulting fees, license/APA compensation, and scientific advisory board compensation from Amylyx. **KF** and **MM** are or were full-time employees of and may have stock option ownership in Amylyx Pharmaceuticals, Inc.

References

1. Salehi M, et al. *J Clin Endocrinol Metab.* 2018;103(8):2815-2826. **2.** McLaughlin T, et al. *J Clin Endocrinol Metab.* 2010;95(4):1851-5. **3.** Craig CM, et al. *Diabetes Obes Metab.* 2018;20(2):352-361 **4.** Sheehan A, et al. *Diabetes Metab Syndr Obes.* 2020;13:4469-4482. **5.** Hazlehurst J, et al. *Endocr Connect.* 2024;13(5):e230285. **6.** Craig CM, et al. *J Clin Endocrinol Metab.* 2021;106(8)e3235-e3248. **7.** Wang X, et al. Int *J Endocrinol.* 2015:651757. **8.** Zilstorff D, et al. *BMC Endocr Disord.* 2024;24:38. **9.** Hallworth J, et al. *J Nutr Metab.* 2017:4823102.

PRESENTED AT ENDO 2025; JULY 12 - 15, 2025; SAN FRANCISCO, CALIFORNIA

© 2025 Amylyx Pharmaceuticals, Inc.