Reduction in Rate of Hypoglycemic Events with Avexitide in Post-Bariatric Hypoglycemia: Results from the Phase 2 and 2B Studies

Colleen Craig, MD[1*]; Tracey McLaughlin, MD, MS[1*]; Zhengyu Yang, PhD[2*]; Kelly Fox, MD[2*]; Ryan Miller, MD[2*]; Helen Margaret Lawler, MD[3*]; Dawn Belt Davis, MD, PhD[4*]; Marilyn Tan, MD, FACE[1*]

[1]Stanford University School of Medicine, Department of Medicine, Stanford, California, USA; [2]Amylyx Pharmaceuticals, Inc., Cambridge, Massachusetts, USA; [3]University of Colorado School of Medicine, Division of Endocrinology, Metabolism, and Diabetes, Department of Medicine, Aurora, Colorado, USA. [4]University of Wisconsin, Division of Endocrinology, Diabetes, and Metabolism, Department of Medicine, Madison, Wisconsin, USA. *Potential conflict of interest may exist. Refer to the Disclosures section.

SUN-627

AMYLYX

BACKGROUND

- Post-bariatric hypoglycemia (PBH) is a chronic condition characterized by hyperinsulinemic hypoglycemia that develops in some individuals who have undergone bariatric surgery[1-6]
- PBH is believed to be caused by changes in hormonal and glycemic patterns, including an excessive glucagon-like peptide-1 (GLP-1) response, as a result of altered nutrient transit post-surgery[1-4]
- PBH can cause debilitating hypoglycemic events associated with neuroglycopenia, including impaired cognition, poor coordination, loss of consciousness, and seizures which can result in a high degree of disability and major disruptions to independent living[5]
- Avexitide is an investigational, first-in-class glucagon-like peptide-1 (GLP-1) receptor antagonist designed to block the effect of excessive GLP-1, targeting a central pathway of PBH pathophysiology to mitigate hypoglycemia by decreasing insulin secretion and stabilizing glucose levels[1-4]
- To date, one Phase 2 randomized, multi-center, placebo-controlled crossover study (PREVENT trial) has been conducted to evaluate the efficacy and safety of avexitide for the treatment of PBH[6]
- In addition, 8 investigator-initiated studies (including a Phase 2b study) have been performed that together demonstrated the effects of avexitide treatment on reducing hypoglycemia in individuals with PBH and those with hyperinsulinemic hypoglycemia following other, non-bariatric gastrointestinal surgeries (e.g., gastrectomy)[3,7-13]

PROPOSED AVEXITIDE MECHANISM OF ACTION

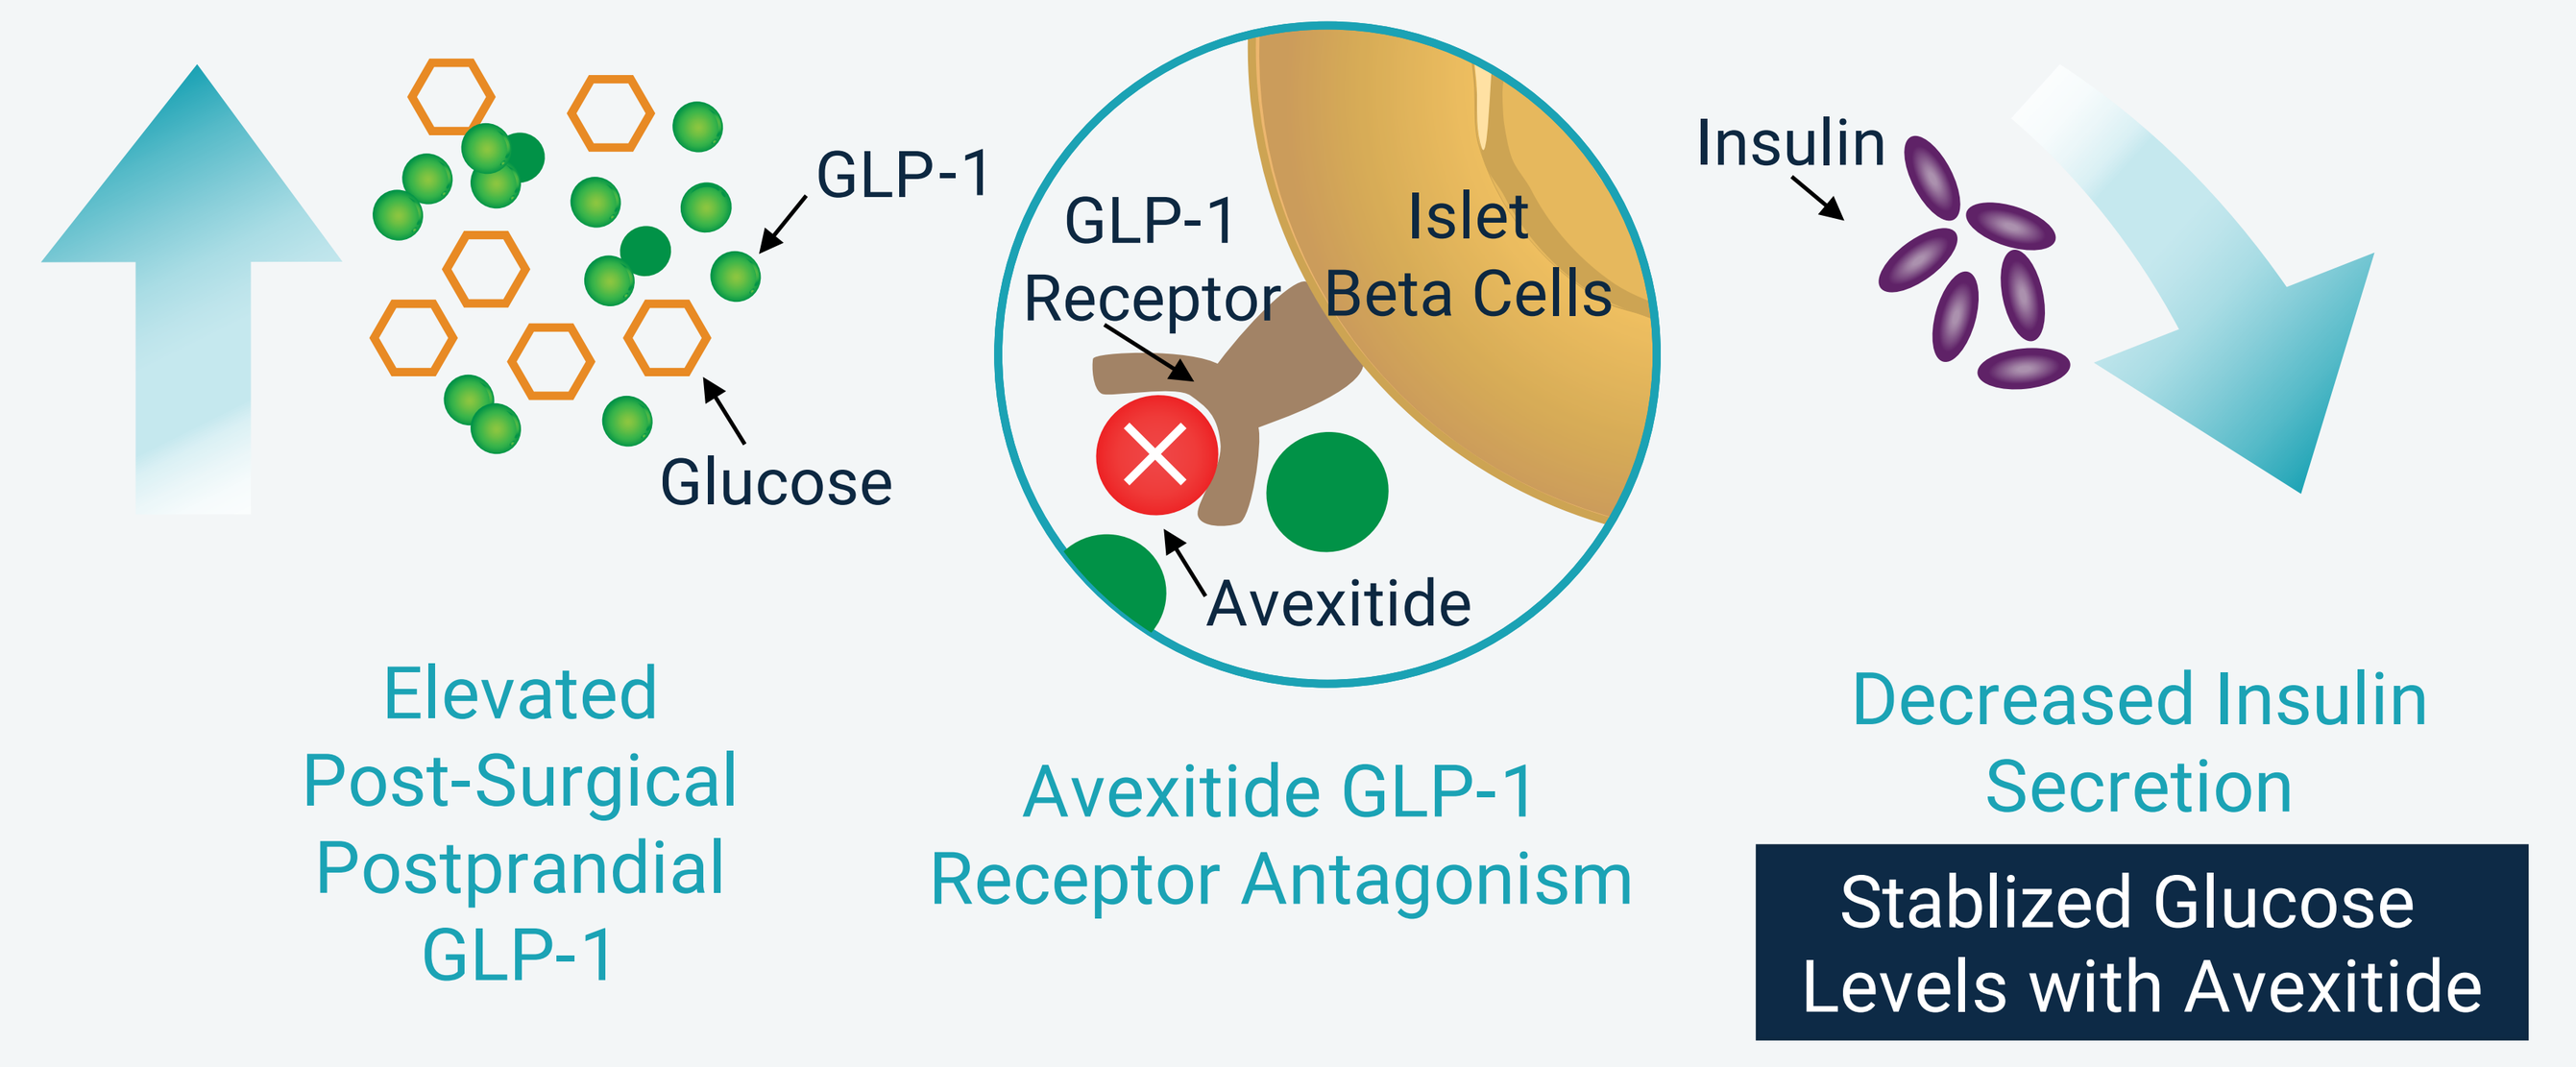
GLP-1
Glucose
GLP-1
Receptor
Islet
Beta Cells
Avexitide
Insulin
Elevated
Post-Surgical
Postprandial
GLP-1
Avexitide GLP-1
Receptor Antagonism
Decreased Insulin
Secretion
Stablized Glucose
Levels with Avexitide

AVEXITIDE (AVX) PHASE 2[6] & 2b[7] CLINICAL TRIAL DESIGNS AND SELECT RESULTS

PHASE 2 PREVENT TRIAL DESIGN – 28-day, placebo-controlled crossover randomized, trial (N = 18[a])

PHASE 2 PHASE 2B TRIAL DESIGN – 28-day, open-label, investigator-initiated, crossover trial (N = 17[a])

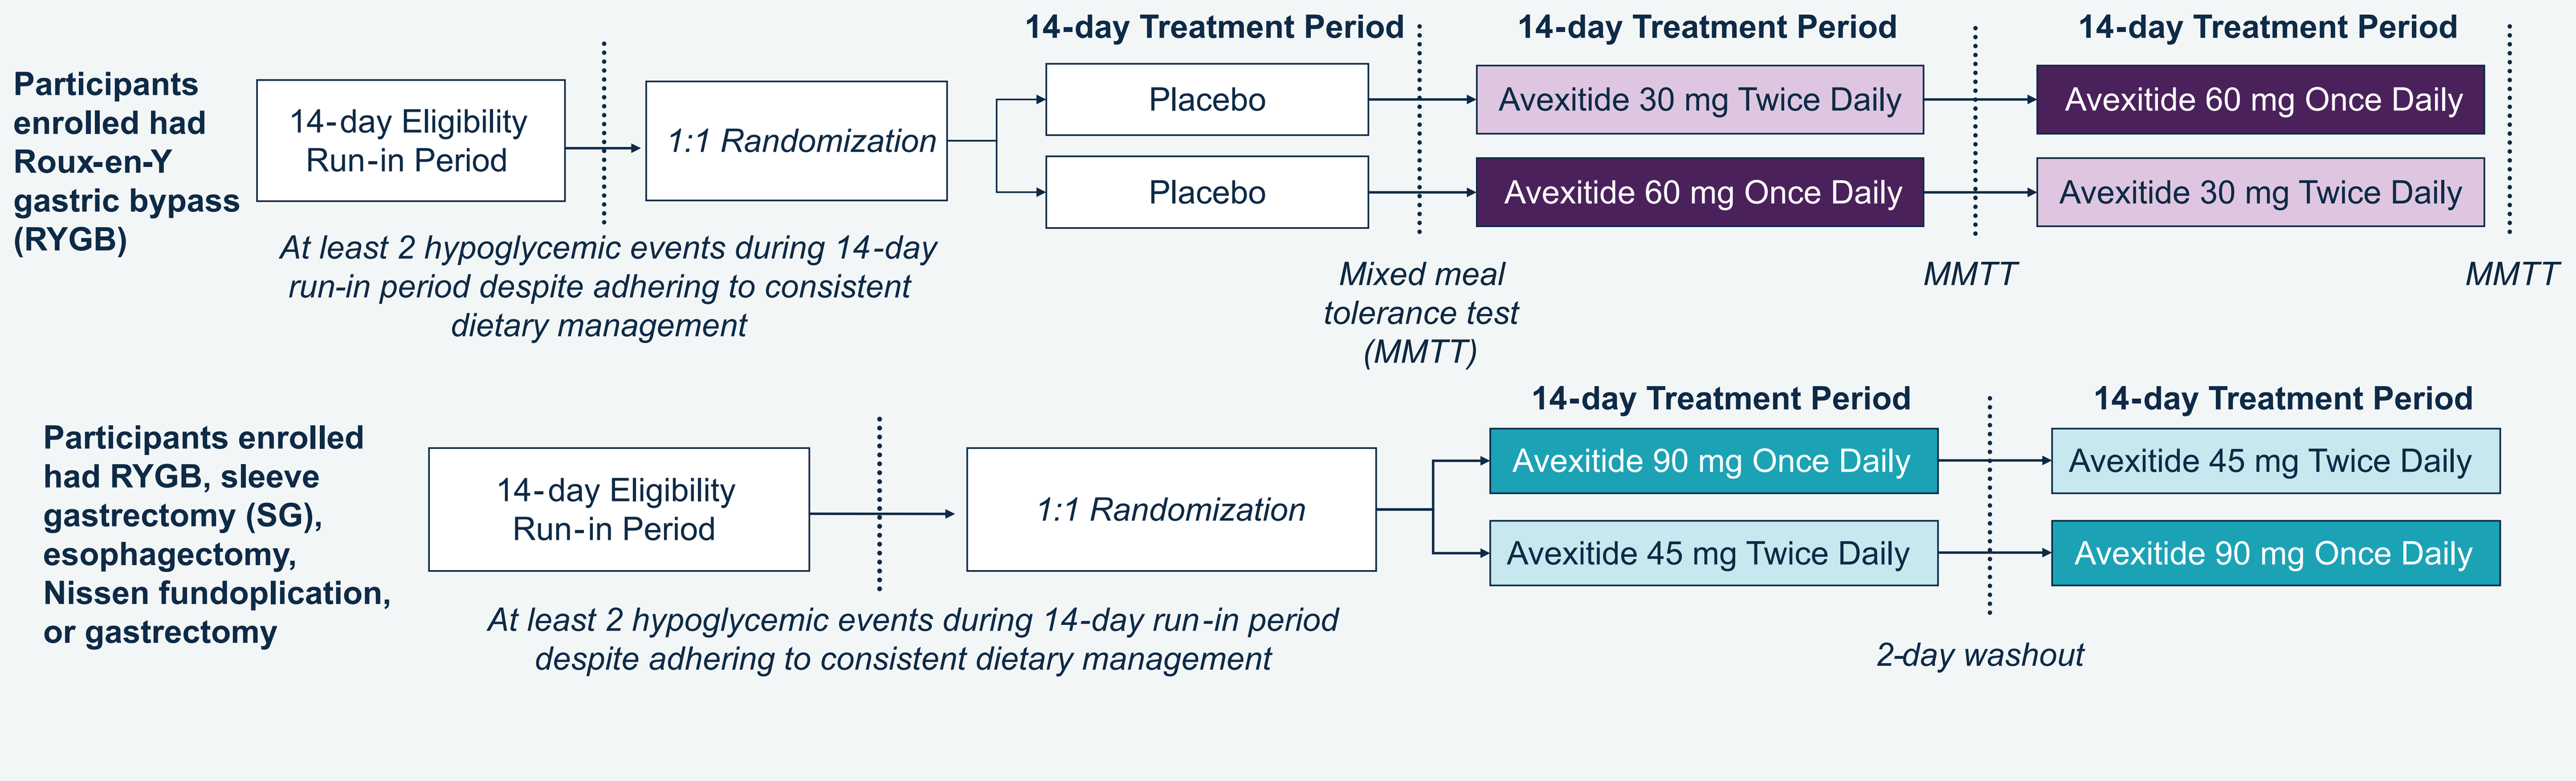
Participants enrolled had Roux-en-Y gastric bypass (RYGB)
14-day Eligibility Run-in Period
1:1 Randomization
14-day Treatment Period
Placebo
Placebo
14-day Treatment Period
Avexitide 30 mg Twice Daily
Avexitide 60 mg Once Daily
14-day Treatment Period
Avexitide 60 mg Once Daily
Avexitide 30 mg Twice Daily
At least 2 hypoglycemic events during 14-day run-in period despite adhering to consistent dietary management
Mixed meal tolerance test (MMTT)
MMTT
MMTT
Participants enrolled had RYGB, sleeve gastrectomy (SG), esophagectomy, Nissen fundoplication, or gastrectomy
14-day Eligibility Run-in Period
1:1 Randomization
14-day Treatment Period
Avexitide 90 mg Once Daily
Avexitide 45 mg Twice Daily
14-day Treatment Period
Avexitide 45 mg Twice Daily
Avexitide 90 mg Once Daily
At least 2 hypoglycemic events during 14-day run-in period despite adhering to consistent dietary management
2-day washout

% Reduction in Rate[b] of Hypoglycemia by Avexitide Dose

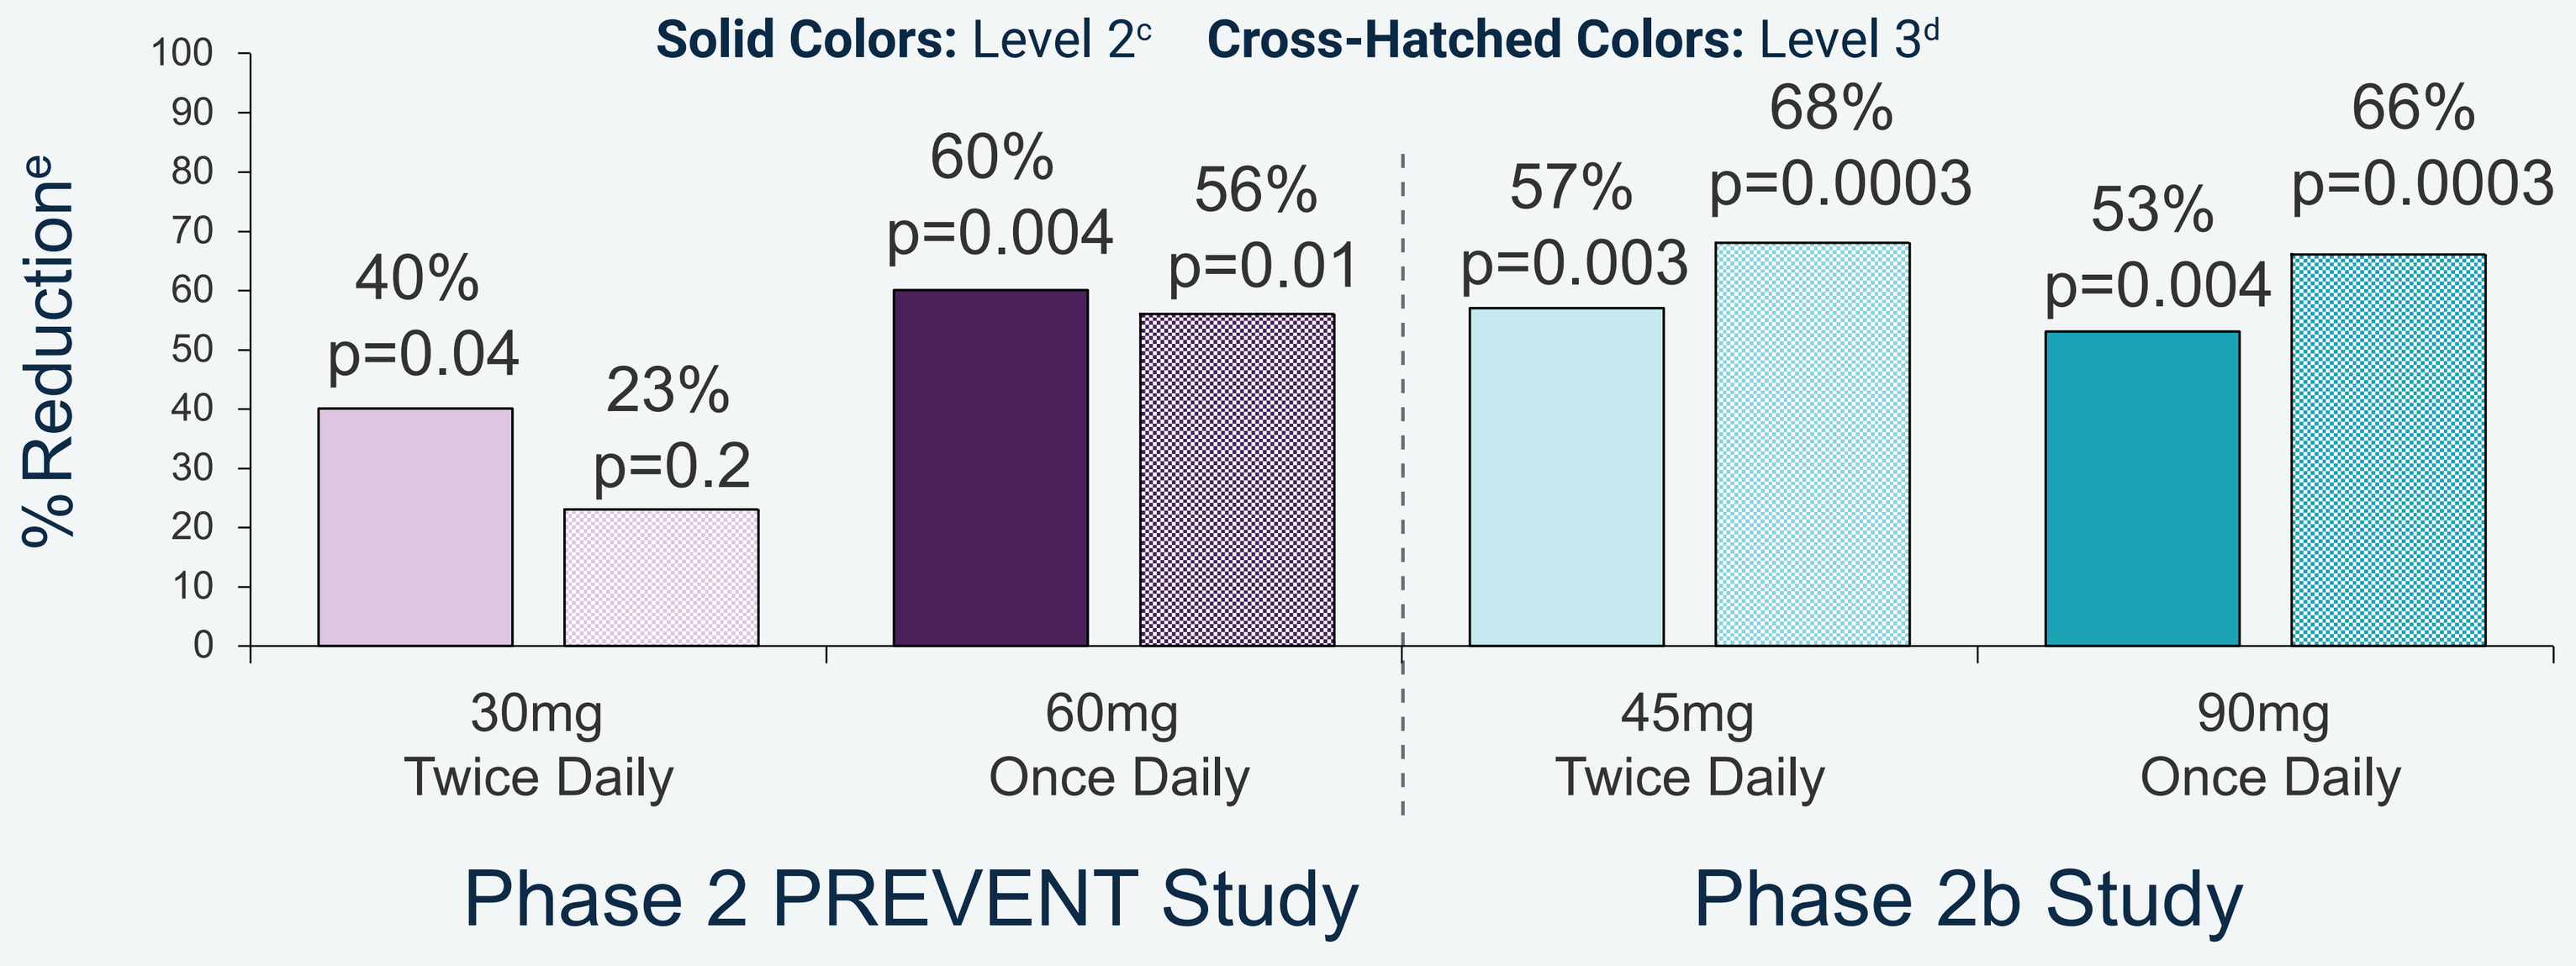
Solid Colors: Level 2[c] Cross-Hatched Colors: Level 3[d]
% Reduction[e]
100
90
80
70
60
50
40
30
20
10
0
40% p=0.04
23% p=0.2
60% p=0.004
56% p=0.01
57% p=0.003
68% p=0.0003
53% p=0.004
66% p=0.0003
30mg Twice Daily
60mg Once Daily
45mg Twice Daily
90mg Once Daily
Phase 2 PREVENT Study
Phase 2b Study

- Avexitide demonstrated statistically significant reductions in hypoglycemic events in Phase 2 and 2b clinical trials
- No clinically relevant increases were observed in fasting or peak postprandial plasma glucose levels
- Adverse events have generally been mild to moderate and transient with no treatment-related serious adverse events and no adverse events necessitating drug discontinuation

[a] *One excluded from the efficacy analysis due to major protocol deviation*
[b] *Rate defined as number of episodes in each treatment period normalized to 14 days*
[c] *Level 2 hypoglycemia: self-monitoring of blood glucose <54 mg/dL*
[d] *Level 3 hypoglycemia: a severe event characterized by altered mental and/or physical functioning that requires assistance from another person for recovery whether an individual receives external assistance or not*
[e] *Compared to placebo in Phase 2 and run-in in Phase 2b*

OBJECTIVE

Given that the phase 3 LUCIDITY trial will further evaluate avexitide in PBH with a primary efficacy outcome of composite reduction of Level 2 and 3 hypoglycemic events, the goal in this exploratory analysis is to understand the composite reduction of Level 2 and 3 hypoglycemic events in prior Phase 2 trials of avexitide in PBH

METHODS

- For each avexitide dose, the rate of composite Level 2 and 3 events[c,d] was calculated as the weekly number of discrete events during the respective treatment periods of the Phase 2 and 2b clinical trials
 - A negative binomial regression model was used, and a least-squares (LS) mean rate ratio (RR) vs placebo (for the Phase 2) and run-in (for the Phase 2b) of hypoglycemia during the treatment period was calculated
 - For the Phase 2b, the model included treatment, treatment sequence, and surgery type stratum as fixed effect, event rate in run-in period as covariate, and offset by duration of treatment period

RESULTS

FIGURE 1. AVX Significantly Reduced Rates of Composite Level 2&3 Hypoglycemia

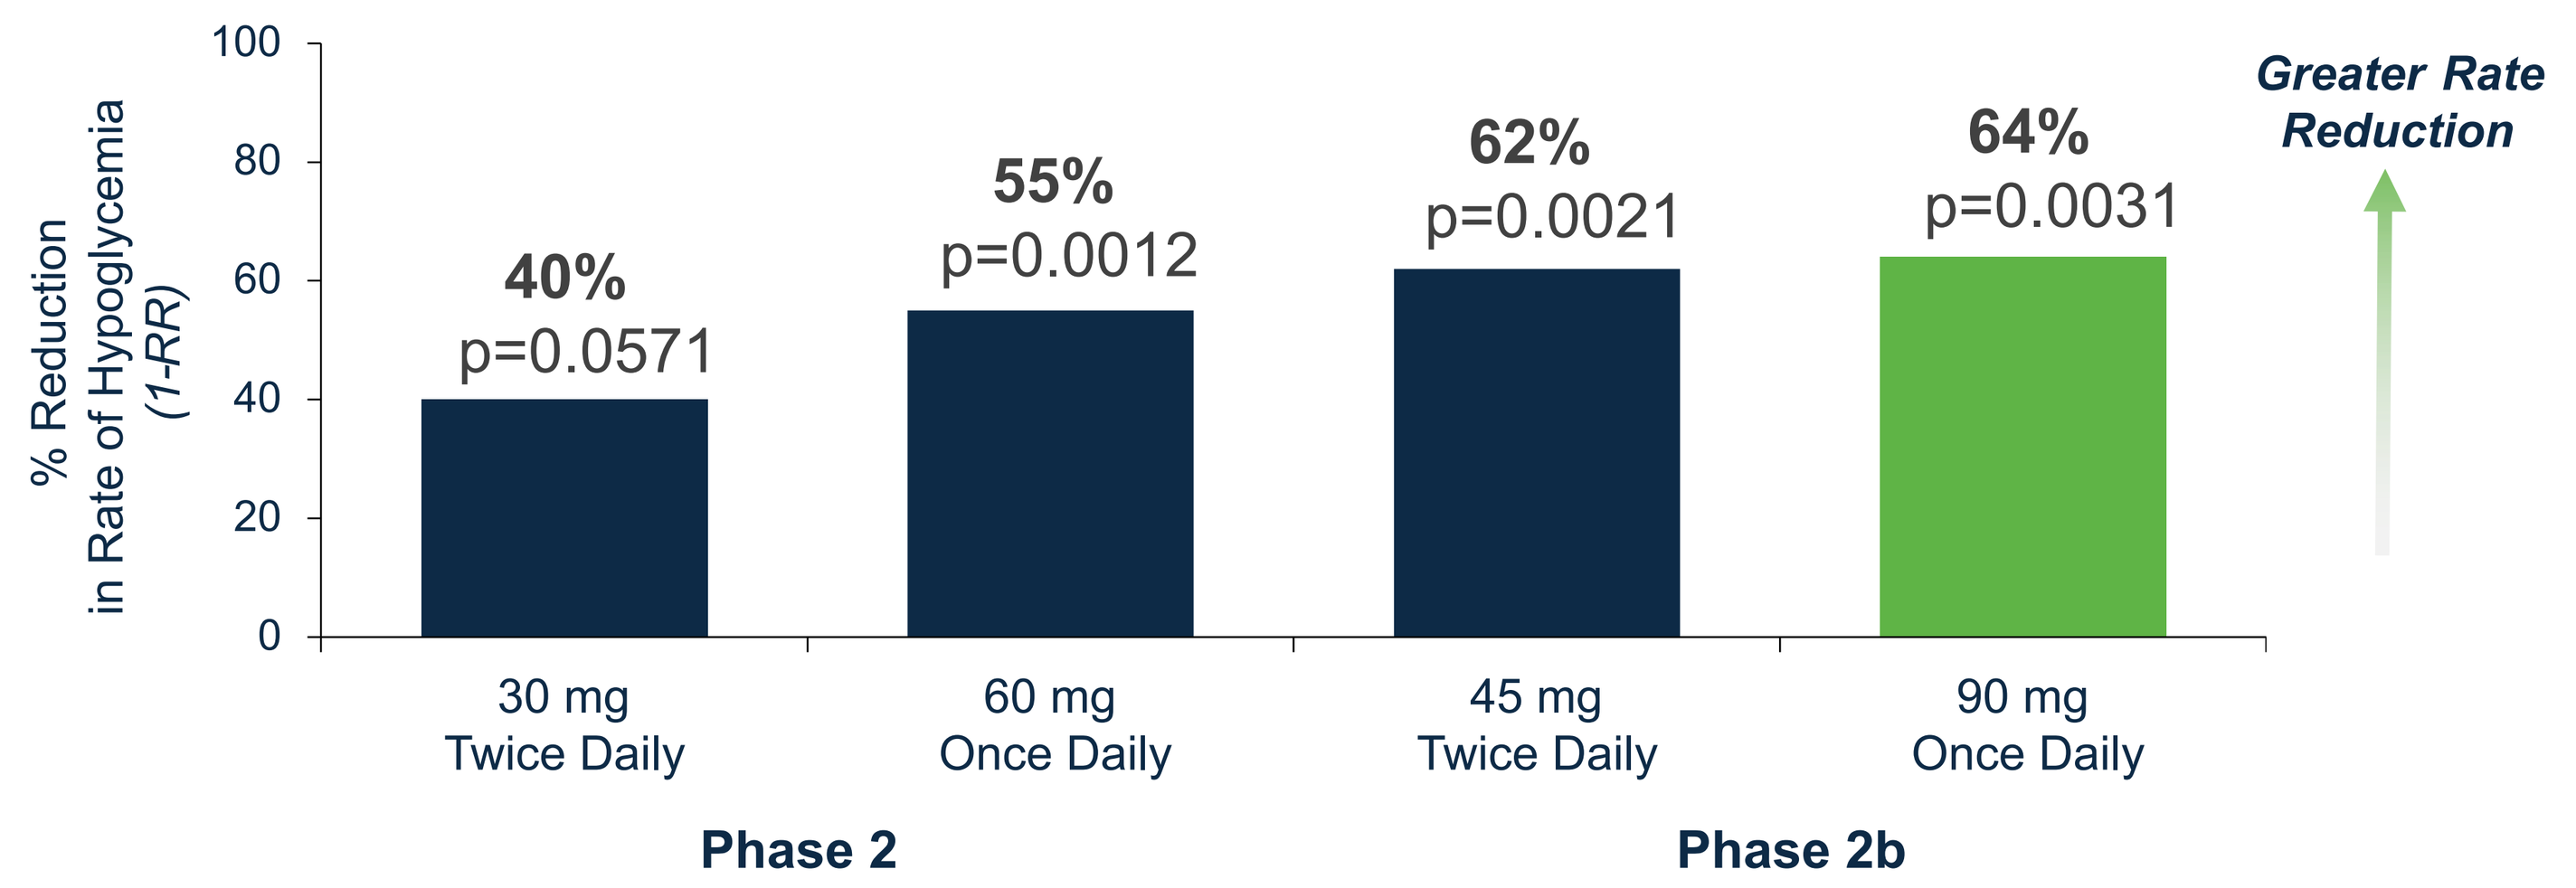
% Reduction in Rate of Hypoglycemia (1-RR)
100
80
60
40
20
0
40% p=0.0571
55% p=0.0012
62% p=0.0021
64% p=0.0031
Greater Rate Reduction
30 mg Twice Daily
60 mg Once Daily
45 mg Twice Daily
90 mg Once Daily
Phase 2
Phase 2b

TABLE 1. Reduced Rates of Composite Level 2 and 3 Hypoglycemia in Phase 2

	Run-In Period n=17	Placebo n=17	Avexitide 30 mg Twice Daily n=17	Avexitide 60 mg Once Daily n=17
Composite Weekly Rate of Level 2&3 Hypoglycemia				
Mean (SD)	1.37 (1.244)	1.47 (1.049)	0.89 (1.101)	0.66 (0.609)
Median	1.04	1.50	0.50	0.50
Rate Ratio *(over placebo of hypoglycemia during treatment period)*				
LS Mean (SE)	N/A	N/A	0.60 (0.260)	0.45 (0.233)
95% CI	N/A	N/A	0.357, 1.016	0.281, 0.717
P-Value	N/A	N/A	0.0571	0.0012

Table 2. Reduced Rates of Composite Level 2 and 3 Hypoglycemia in Phase 2B

	Run-In Period n=16	Avexitide 45 mg Twice Daily n=16	Avexitide 90 mg Once Daily n=16
Composite Weekly Rate of Level 2&3 Hypoglycemia			
Mean (SD)	1.39 (1.908)	0.51 (0.719)	0.59 (1.604)
Median	0.93	0.25	0
Rate Ratio *(over run-in of hypoglycemia during treatment period)*			
LS Mean (SE)	N/A	0.38 (0.298)	0.36 (0.325)
95% CI	N/A	0.206, 0.687	0.187, 0.694
P-Value	N/A	0.0021	0.0031

Abbreviations: CI, confidence interval; SD, standard deviation; SE, standard error

CONCLUSIONS

- Avexitide significantly reduced rates of composite Level 2 and 3 hypoglycemia in its Phase 2 and 2b trials
- The 90 mg once daily dose, which is being further evaluated in the Phase 3 LUCIDITY trial, led to a 64% reduction in the composite rate of Level 2 and 3 events (p=0.0031)
 - With 90 mg once daily, mean and median number of events per week were 0.59 and 0, respectively, vs 1.39 and 0.93 during the run-in period
- The Phase 3 LUCIDITY trial is currently recruiting in the United States with topline data anticipated in 1H 2026

lucidity

SCAN TO LEARN MORE ABOUT THE PHASE 3 LUCIDITY TRIAL

CONTACT INFO: KELLY FOX, MD
Kelly_Fox@amylyx.com

Avexitide is an investigational drug and has not been approved for use by any health authority (e.g., the FDA and EMA).

Acknowledgements

The authors would like to thank the Phase 2 and 2b trial participants and their families. The Phase 2 and 2b studies were sponsored by Eiger BioPharmaceuticals, Inc. This analysis was sponsored by Amylyx Pharmaceuticals, Inc.

Disclosures

CC reports consulting fees, license/APA compensation, and compensation (including travel-related) for participation in a scientific advisory board from Amylyx. **ZY, KF**, and **RM** are or were full-time employees of and may have stock option ownership in Amylyx Pharmaceuticals, Inc. TM reports consulting fees from Amylyx, patents licensed to Amylyx, and compensation (including travel-related) for participation in a scientific advisory board from Amylyx. **HML** reports compensation (including travel-related) for participation in a scientific advisory board from Amylyx and reports consulting fees from Vogenx. DBD reports compensation (including travel-related) for participation in a scientific advisory board from Amylyx and reports reimbursement of travel from Recordati. **MT** reports consulting fees and scientific advisory board and travel compensation from Amylyx.

References

1. Salehi M, et al. J Clin Endocrinol Metab. 2018;103(8):2815-2826. **2.** McLaughlin T, et al. J Clin Endocrinol Metab. 2010;95(4):1851-5. **3.** Craig CM, et al. Diabetes Obes Metab. 2018;20(2):352-361. **4.** Sheehan A, et al. Diabetes Metab Syndr Obes. 2020;13:4469-4482. **5.** Hazlehurst J, et al. Endocr Connect. 2024;13(5):e230285. **6.** Craig CM, et al. J Clin Endocrinol Metab. 2021;106(8)e3235-e3248. **7.** Tan M. Oral presentation at: ENDO 2022; June 11-14, 2022; Atlanta, Georgia. **8.** Salehi M, et al. Diabetes. 2011;60(9):2308-2314. **9.** Salehi M, et al. Gastroenterology. 2014;146(3):669-680. **10.** Larraufie P, et al. Cell Rep. 2019;26(6):1399-1408. **11.** Calabria AC, et al. Horm Res Paediatr. 2016;85(2):140-146. **12.** Craig CM, et al. Diabetologia. 2017;60(3):531-540. 13. Tan M, et al. Diabetes Obes Metab. 2020;22(8):1406-1416.

Presented at ENDO 2025; July 12 - 15, 2025; San Francisco, CA
© 2025 Amylyx Pharmaceuticals, Inc.