Novel Neuroprotective Strategies in Human Neuron Models of ALS/FTD:
Evaluating Antisense Oligonucleotide Therapies, including AMX0114

Michael Bekier[1], Christopher Altheim[1], Evan Mizerak[2], Sami J Barmada[1]

[1]University of Michigan School of Medicine, Department of Neurology, Ann Arbor, MI; [2]Amylyx Pharmaceuticals, Inc., Cambridge, MA

BACKGROUND

- Neurodegeneration in amyotrophic lateral sclerosis (ALS) and frontotemporal dementia (FTD) shares key molecular events with axonal degeneration (also known as Wallerian degeneration)[1,2]
 - Specifically, neuronal injury results in cytoplasmic mislocalization of TAR DNA binding protein 43 (TDP43), with subsequent return of TDP43 to the nucleus upon recovery[3]
 - Identical injury paradigms also upregulate cysteine protease calpain-2 (CAPN2), in part through the disruption of calcium homeostasis, and promote the systematic dismantling of neuronal processes and soma proximal to the site of injury[4-6]
- Several lines of evidence have implicated calpain-2 specifically in the pathogenesis of ALS including:
 - Elevated calpain-2 messenger RNA (mRNA) in muscle samples and calpain-specific TDP43 cleavage product concentrations in postmortem ALS spinal cord samples[7,8]
 - Therapeutic benefit of calpain-2 activity modulation in animal models of ALS[9]
 - The role of calpain-2 in cleaving neurofilament, a broadly researched biomarker in ALS[10]
- Amylyx Pharmaceuticals has developed antisense oligonucleotides (ASOs) targeting the gene encoding calpain-2 *(CAPN2)* including AMX0114, the lead candidate, and ASO39, a backup candidate
- AMX0114 has been shown to achieve potent, durable knockdown of *CAPN2* mRNA and calpain-2 protein and exhibit functional efficacy in multiple neuropathy models[11]

OBJECTIVES

- To determine whether genetic strategies targeting CAPN2 prevent neurodegeneration in ALS/FTD models, providing opportunities for compensatory responses and neuronal regeneration

METHODS

- To model ALS/FTD in a high-throughput fashion, and to study the dynamics of TDP43 in living neurons, we labeled endogenous TDP43 in induced pluripotent stem cells (iPSCs) with the photoconvertible protein Dendra2[12]. The homologous recombination vector for this manipulation included either a wild type (WT) backbone, or one that harbored a pathogenic mutation (M337V), thereby creating an isogenic pair of iPSCs expressing either TDP43(WT) or TDP43(M337V)[13].

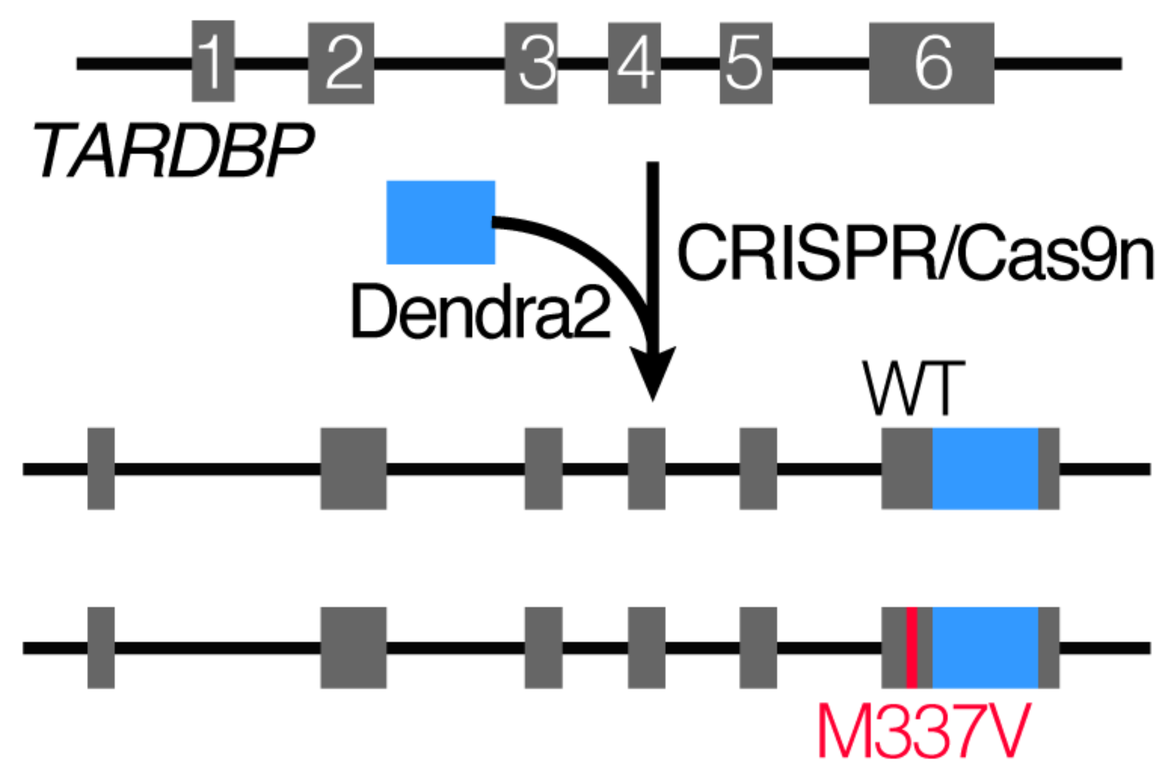
1
2
3
4
5
6
TARDBP
Dendra2
CRISPR/Cas9n
WT
M337V

- To ensure robust iPSC differentiation, we integrated customized cassettes into the *CLYBL* safe harbor locus of iPSCs, allowing for doxycycline (dox)-inducible expression of the transcription factors NGN1 and NGN2 (for glutamatergic cortical iNeurons[12, 14]) or ISL1, NGN2, and LHX3 (for cholinergic spinal iMotorNeurons[12, 15]). In each case, the addition of dox results in the rapid and reproducible differentiation of iPSCs into iNeurons or iMotorNeurons.

RESULTS

Application of genetically-encoded death indicators to assess survival

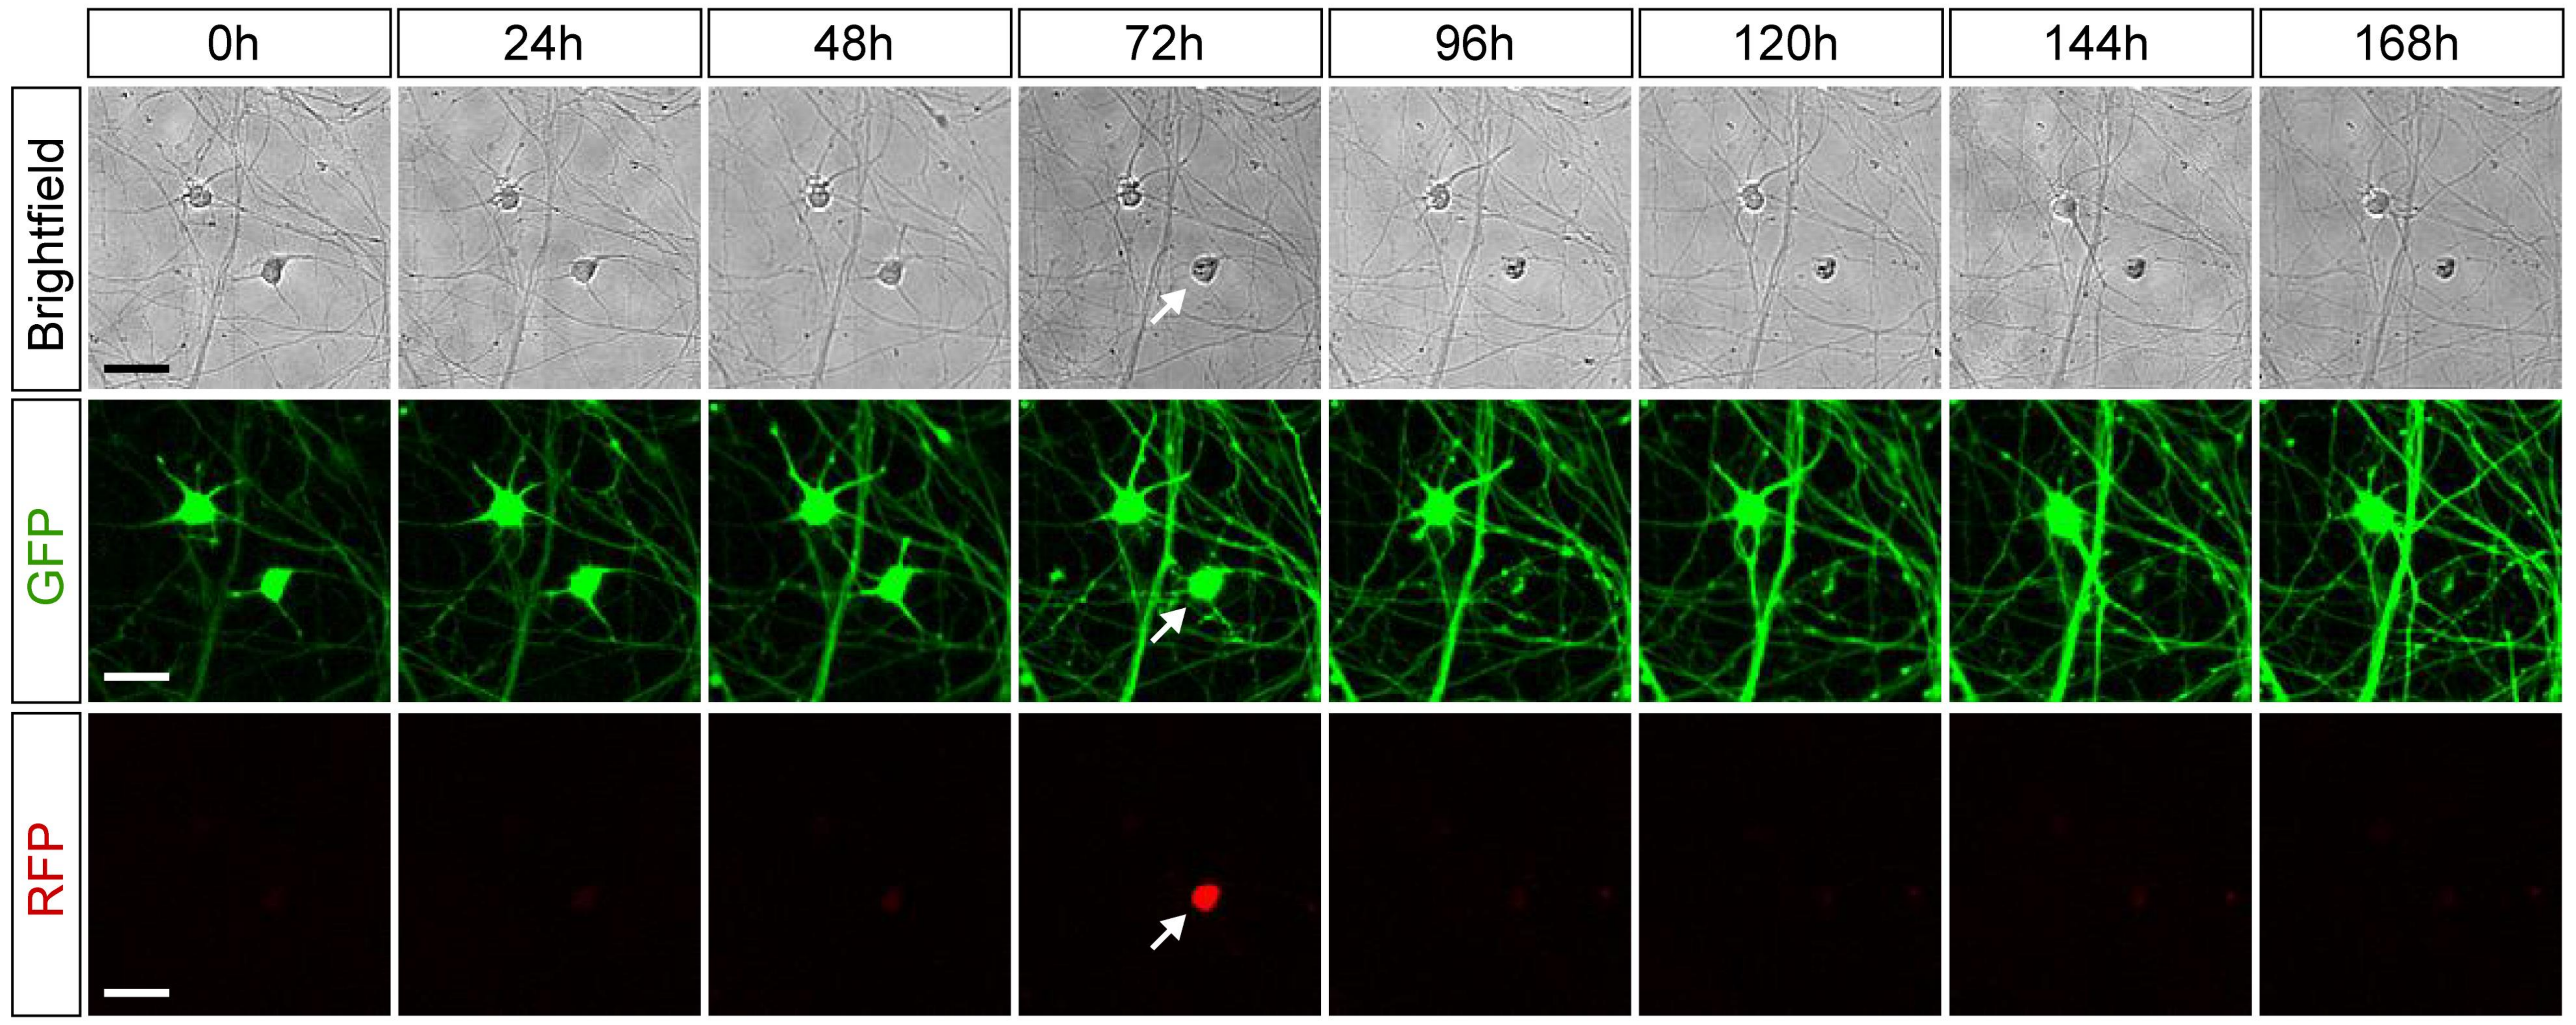
0h
24h
48h
72h
96h
120h
144h
168h
Brightfield
GFP
RFP

Figure 1: Automated imaging and detection of cell death. iPSC-derived neurons (iNeurons) expressing GFP (a cell marker) and red-shifted genetically-encoded death indicator (rGEDI) imaged at regular intervals via longitudinal fluorescence microscopy. Thousands of individual cells are imaged simultaneously in brightfield as well as the GFP and RFP channels. Cell death is marked by red fluorescence (arrow). Scale bar, 10µm. RFP, red fluorescent protein. GFP, green fluorescent protein.

Dose-dependent improvement in survival with AMX0114 and ASO39

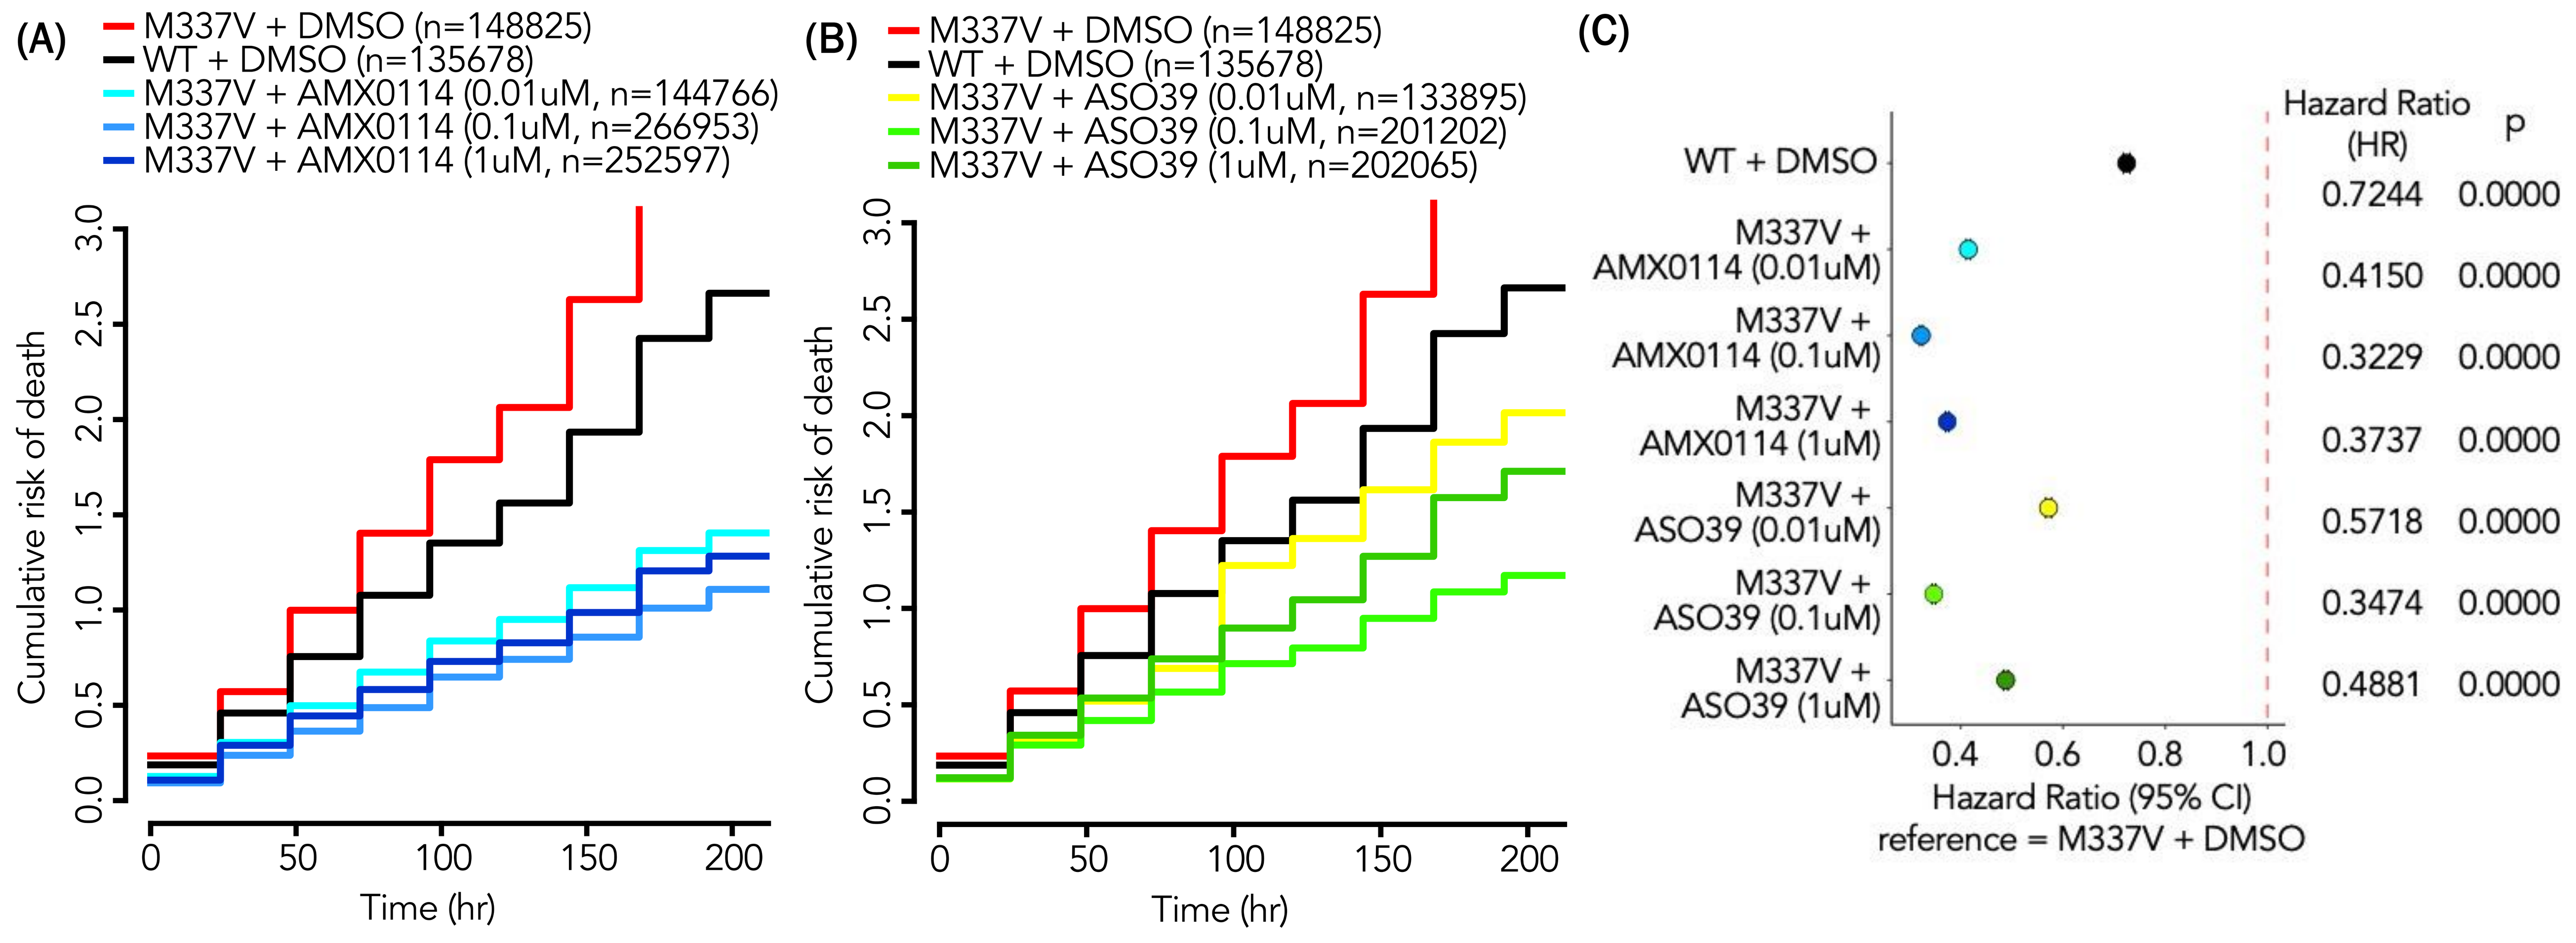
(A)
M337V + DMSO (n=148825)
WT + DMSO (n=135678)
M337V + AMX0114 (0.01uM, n=144766)
M337V + AMX0114 (0.1uM, n=266953)
M337V + AMX0114 (1uM, n=252597)
Cumulative risk of death
Time (hr)
(B)
M337V + DMSO (n=148825)
WT + DMSO (n=135678)
M337V + ASO39 (0.01uM, n=133895)
M337V + ASO39 (0.1uM, n=201202)
M337V + ASO39 (1uM, n=202065)
Cumulative risk of death
Time (hr)
(C)
Hazard Ratio (HR)
p
WT + DMSO 0.7244 0.0000
M337V + AMX0114 (0.01uM) 0.4150 0.0000
M337V + AMX0114 (0.1uM) 0.3229 0.0000
M337V + AMX0114 (1uM) 0.3737 0.0000
M337V + ASO39 (0.01uM) 0.5718 0.0000
M337V + ASO39 (0.1uM) 0.3474 0.0000
M337V + ASO39 (1uM) 0.4881 0.0000
Hazard Ratio (95% CI)
reference = M337V + DMSO

Figure 2: Survival analysis of wild type (WT) and mutant TDP43(M337V) iNeurons treated with AMX0114, Amylyx Pharmaceuticals' lead candidate, and ASO39, a backup candidate. (A) Cumulative risk of death plot for TDP43(WT) and TDP43(M337V) human iNeurons treated with AMX0114 (0.01-1µM). (B) Cumulative risk of death plot for TDP43(WT) and TDP43(M337V) human iNeurons treated with ASO39 (0.01-1µM). (C) Forest plot illustrating hazard ratio (HR) for iNeurons treated with AMX0114 and ASO39, in comparison to TDP43(M337V) iNeurons treated with vehicle control (DMSO; dashed red line). HRs and p values calculated using Cox proportional hazards analysis. n=number of neurons. Data integrated from 6 technical and 3 biological replicates/condition. CI, confidence interval. DMSO, dimethyl sulfoxide.

CONCLUSIONS

- Previous studies have shown efficacious and robust target mRNA and protein knockdown following treatment with CAPN2-targeting ASOs AMX0114 and ASO39[11]
- Prospective imaging and survival analysis demonstrates that this robust knockdown with AMX0114 and ASO39 translates into dose-dependent improvement in the survival of human iPSC-derived neurons in culture

FUTURE DIRECTIONS

- Ongoing analyses are focused on whether ASOs targeting CAPN2 affect the stability of calpain-2 substrates such as alpha-II spectrin
- Additional studies will uncover the impact of CAPN2-targeting ASOs on TDP43 localization and splicing activity. In particular, we are examining characteristic cryptic splicing events in STMN2 and UNC13 regulated by TDP43
- We expect that neuroprotection from CAPN2-targeting ASOs will also extend to iMotorNeurons, which we can explore further
- AMX0114 is currently in investigational new drug (IND)–enabling studies to support clinical study initiation
- IND-enabling studies are scheduled to complete in 2024 which may allow for initiation of a first-in-human study during 2024

AMX0114 is an investigational drug and not approved for use by any health authority.

Disclosures:

- EM is a full-time employee of and may have stock ownership in Amylyx Pharmaceuticals, Inc.
- This study is sponsored by Amylyx Pharmaceuticals, Inc.

References

1. Coleman M and Hoke A. *Nat Rev Neurosci.* 2022;21(4):183-196.
2. Stubbs K, et al. *Brain Commun.* 2023; 5(2):fcad052.
3. Jo M, et al. *Exp Mol Med.* 2020;52(10):1652-1662.
4. Chang B, et al. *Eur J Neurosci.* 2016;44(4):2040-8.
5. Ma M, et al. *Neurobiol Dis.* 2013;56:34-46.
6. Wang Y, et al. *Cells.* 2020; 9(12):2698.
7. Ueyama H, et al. *J Neurol Sci*. 1998;155(2):163-169.
8. Yamashita T, et al. *Nat Commun*. 2012;3:1307.
9. Rao MV, et al. *J Neurochem*. 2016;137(2):253-265.
10. Ma M. *Neurobiol Dis*. 2013;60:61-79.
11. Mizerak E, et al. Poster presented at TIDES USA 2023; May 7-10, 2023; San Diego, CA and virtual.
12. Chua J, et al. *Curr Biol.* 2022;32(12):2581-2595.e6
13. McMillan M, Gomez N, et al. *Mol Cell.* 2023;83(2):219-236.e7.
14. Weskamp K, et al. *J Clin Inv.* 2020; 130(3):1139-1155.
15. Safren N, et al. *J Biol Chem.* 2021;297(3):101003.

Presented at the 22nd Annual NEALS Meeting; October 4-6, 2023; Clearwater Beach, Florida